HORIZON (OP-106): Melflufen Plus Dexamethasone in Relapsed/Refractory Multiple Myeloma Refractory to Pomalidomide and/or an Anti-CD38 Monoclonal Antibody – Primary and Subgroup Analysis

Poster EP945

Paul G. Richardson[1]; Albert Oriol[2]; Alessandra Larocca[3]; Joan Bladé[4]; Michele Cavo[5]; Paula Rodriguez-Otero[6]; Xavier Leleu[7]; Maxim Norkin[8]; Omar Nadeem[1]; John W. Hiemenz[9]; Hani Hassoun[10]; Cyrille Touzeau[11-13]; Adrián Alegre[14]; Agne Paner[15]; Christopher Maisel[16]; Amitabha Mazumder[17]; Anastasios Raptis[18]; Noemí Puig[19]; Johan Harmenberg[20]; Catriona Byrne[20]; Sara Thuresson[20]; Jakob Obermüller[20]; Maria-Victoria Mateos[19]

[1]Dana-Farber Cancer Institute, Harvard Medical School, Boston, MA, USA; [2]Hospital Germans Trias i Pujol, Badalona, Spain; [3]A.O.U. Città della Salute e della Scienza di Torino - S.C. Ematologia U, Torino, Italy; [4]Hospital Clínica de Barcelona - Servicio de Onco-Hematología, Barcelona, Spain; [5]Policlinico S. Orsola Malpighi, Bologna, Italy; [6]Clínica Universidad de Navarra, Pamplona, Spain; [7]CHU de Poitiers, Poitiers, France; [8]University of Florida Health Cancer Center, Gainesville, FL, USA; [9]Division of Hematology Oncology, Department of Medicine, University of Florida, Gainesville, FL, USA; [10]Myeloma Service, Department of Medicine, Memorial Sloan Kettering Cancer Center, New York, NY, USA; [11]Centre de Recherche en Cancérologie et Immunologie Nantes-Angers (CRCINA), Institut National de la Santé et de la Recherche Médicale (INSERM), Centre National de la Recherche Scientifique (CNRS), Université d'Angers, Université de Nantes, Nantes, France; [12]Site de Recherche Intégrée sur le Cancer (SIRIC), Imaging and Longitudinal Investigations to Ameliorate Decision-making (ILIAD), Nantes, France; [13]Service d'hématologie Clinique, Centre Hospitalier Universitaire, Place Alexis Ricordeau, Nantes, France; [14]Hospital Universitario La Princesa, Madrid, Spain; [15]Rush University Medical Center, Chicago, IL, USA; [16]Baylor Scott & White Charles A. Sammons Cancer Center, Dallas, TX, USA; [17]The Oncology Institute of Hope and Innovation, Glendale, CA, USA; [18]Division of Hematology-Oncology, Department of Medicine, University of Pittsburgh School of Medicine, Pittsburgh, PA, USA; [19]Hospital Clinico Universitario de Salamanca, Salamanca, Spain; [20]Oncopeptides AB, Stockholm, Sweden

BACKGROUND

- Despite advances in therapy, outcomes remain poor for patients with relapsed/refractory multiple myeloma (RRMM)[1,2]
- Treatment choice after relapse generally depends on patient characteristics, prior therapy received, and prior response to therapy[3,4]
- Although class switching is generally prioritized, this is becoming increasingly hard to achieve because patients are being exposed to novel combinations in earlier lines of therapy, resulting in disease that is resistant to multiple drug classes[3-5]
- Melphalan flufenamide (melflufen) is a fist-in-class anticancer peptide-drug conjugate that rapidly delivers an alkylating payload into tumor cells (**Figure 1**)[6-10]
- The efficacy and safety of melflufen plus dexamethasone (dex) was initially demonstrated in O-12-M1, a phase 1/2, dose-finding study in patients with RRMM who had received a median of 4 prior lines of therapy (including lenalidomide and bortezomib), and had disease refractory to their last line of therapy[11]
 - Among 45 patients who received melflufen plus dex, overall response rate (ORR) was 31%, median duration of response (DOR) was 8.4 months, and median progression-free survival (PFS) was 5.7 months
 - The safety profile of melflufen plus dex was primarily characterized by clinically manageable hematologic adverse events (AEs) and a low frequency of nonhematologic AEs

Figure 1. Melflufen Mechanism of Action

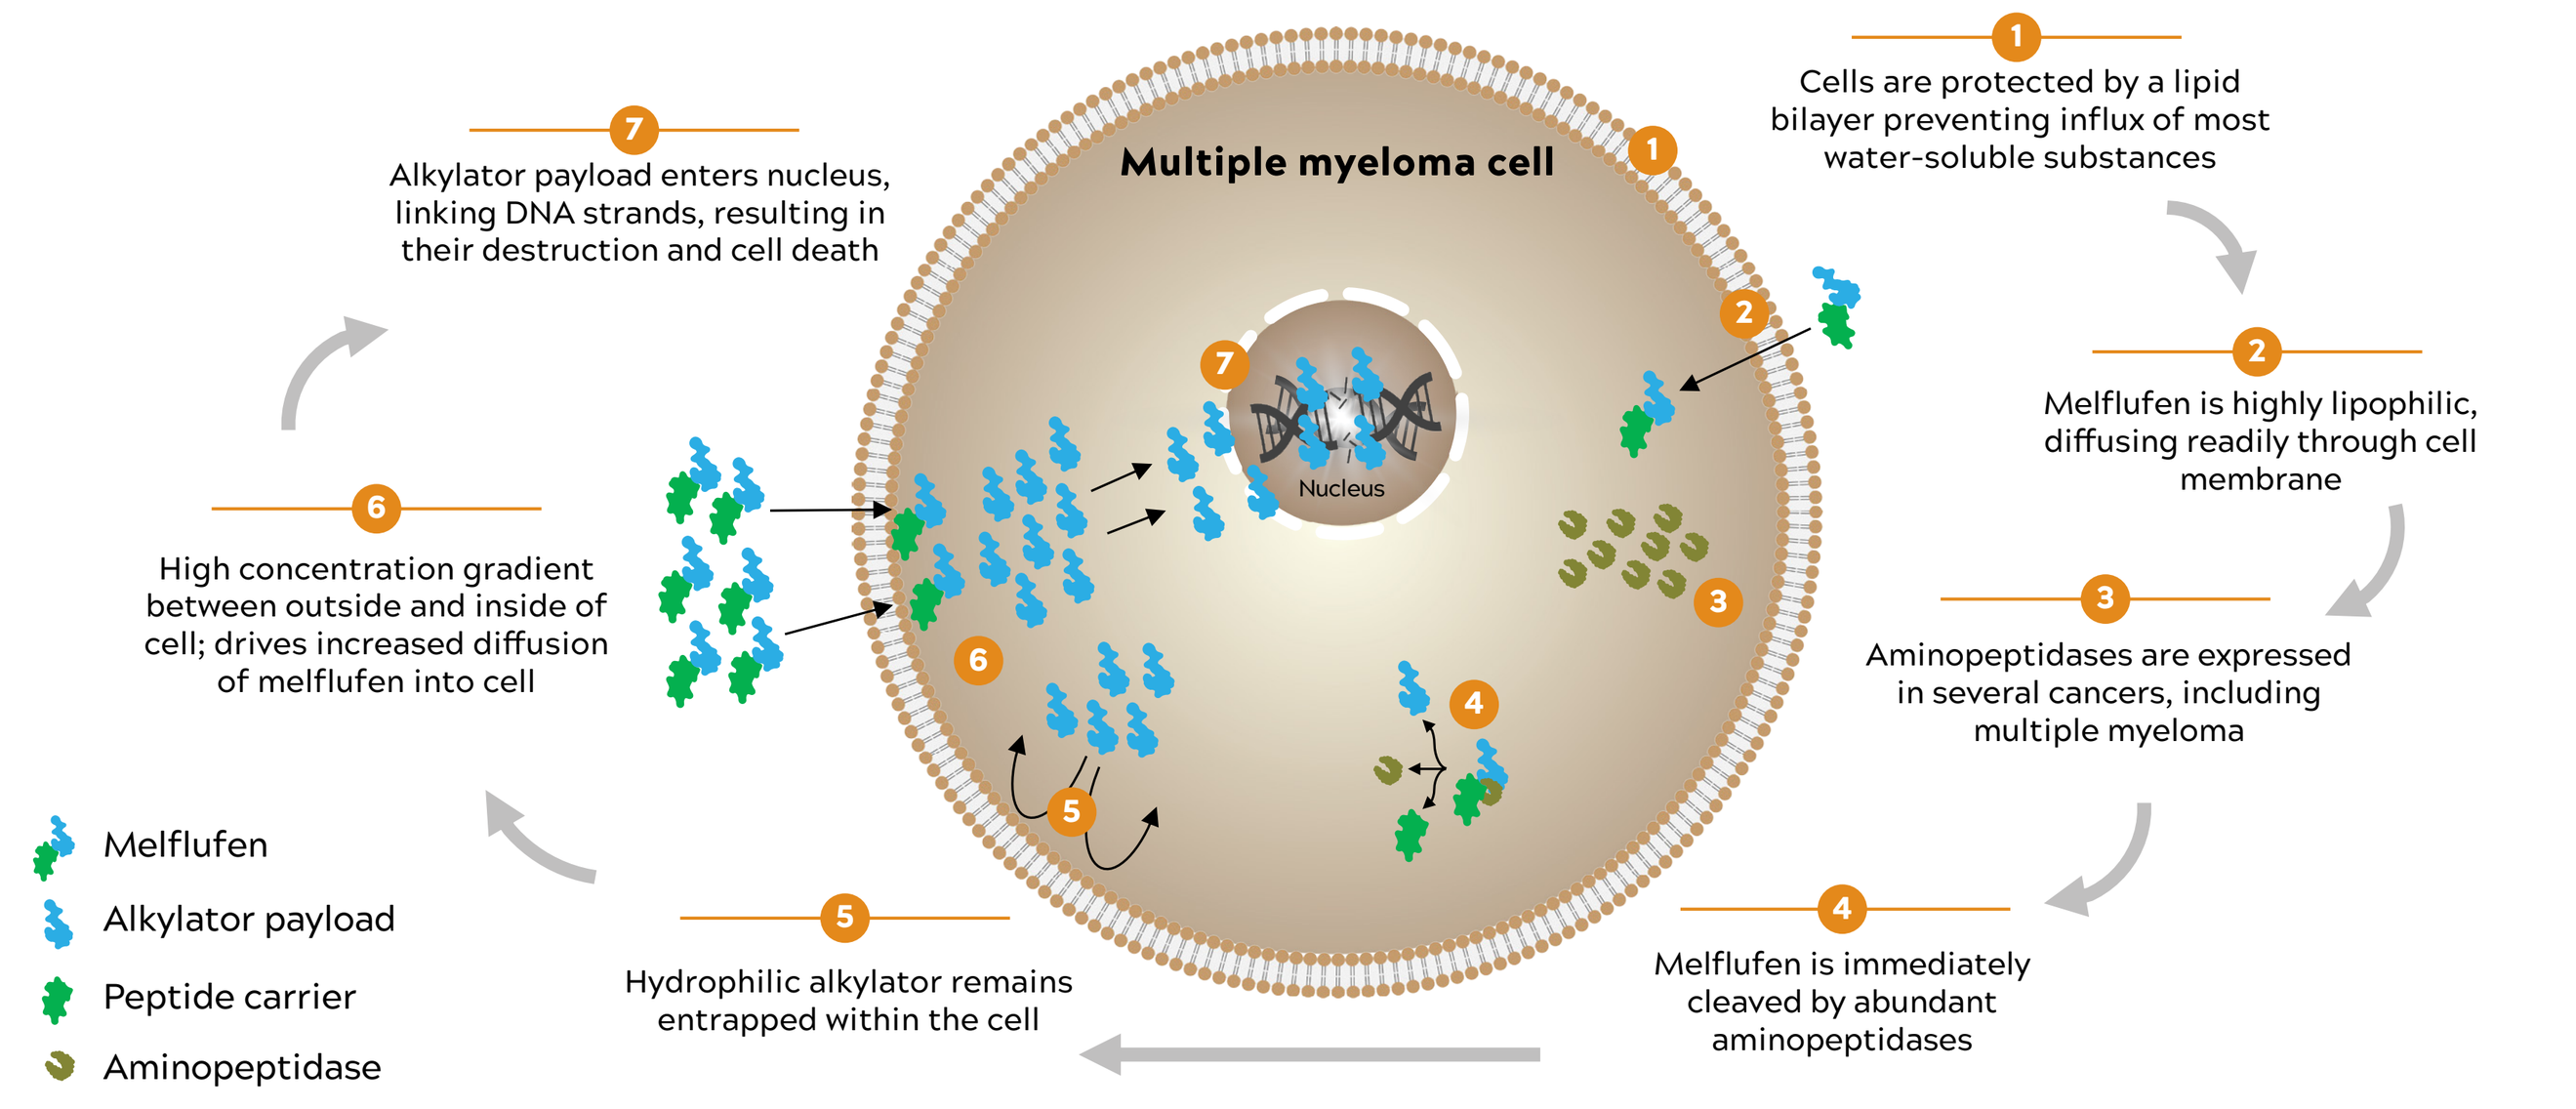

Multiple myeloma cell
Nucleus
1 Cells are protected by a lipid bilayer preventing influx of most water-soluble substances
2 Melflufen is highly lipophilic, diffusing readily through cell membrane
3 Aminopeptidases are expressed in several cancers, including multiple myeloma
4 Melflufen is immediately cleaved by abundant aminopeptidases
5 Hydrophilic alkylator remains entrapped within the cell
6 High concentration gradient between outside and inside of cell; drives increased diffusion of melflufen into cell
7 Alkylator payload enters nucleus, linking DNA strands, resulting in their destruction and cell death
Melflufen
Alkylator payload
Peptide carrier
Aminopeptidase

OBJECTIVE

- To evaluate the efficacy and safety of melflufen plus dex in patients with RRMM in the pivotal, single-arm, multicenter, phase 2 HORIZON study

METHODS

- HORIZON is a pivotal, single-arm, multicenter, phase 2 study of melflufen plus dex in patients with RRMM who had received ≥ 2 prior lines of therapy and were refractory to pomalidomide (pom) and/or an anti-CD38 monoclonal antibody (mAb; **Figure 2**)
 - Treatment: melflufen 40 mg (day 1) in combination with dex 40 mg (days 1, 8, 15, and 22; 20 mg in patients aged ≥75 years) during each 28-day treatment cycle until disease progression or unacceptable toxicity
- The intention-to-treat (ITT) population was used for all analyses. Subgroups of special interest included patients with triple-class–refractory disease (refractory to or intolerant of ≥1 IMiD + ≥1 proteasome inhibitor [PI] + ≥1 anti-CD30 mAb) and patients with extramedullary disease (EMD)

Figure 2. Phase 2 HORIZON Study Design (NCT02963493)

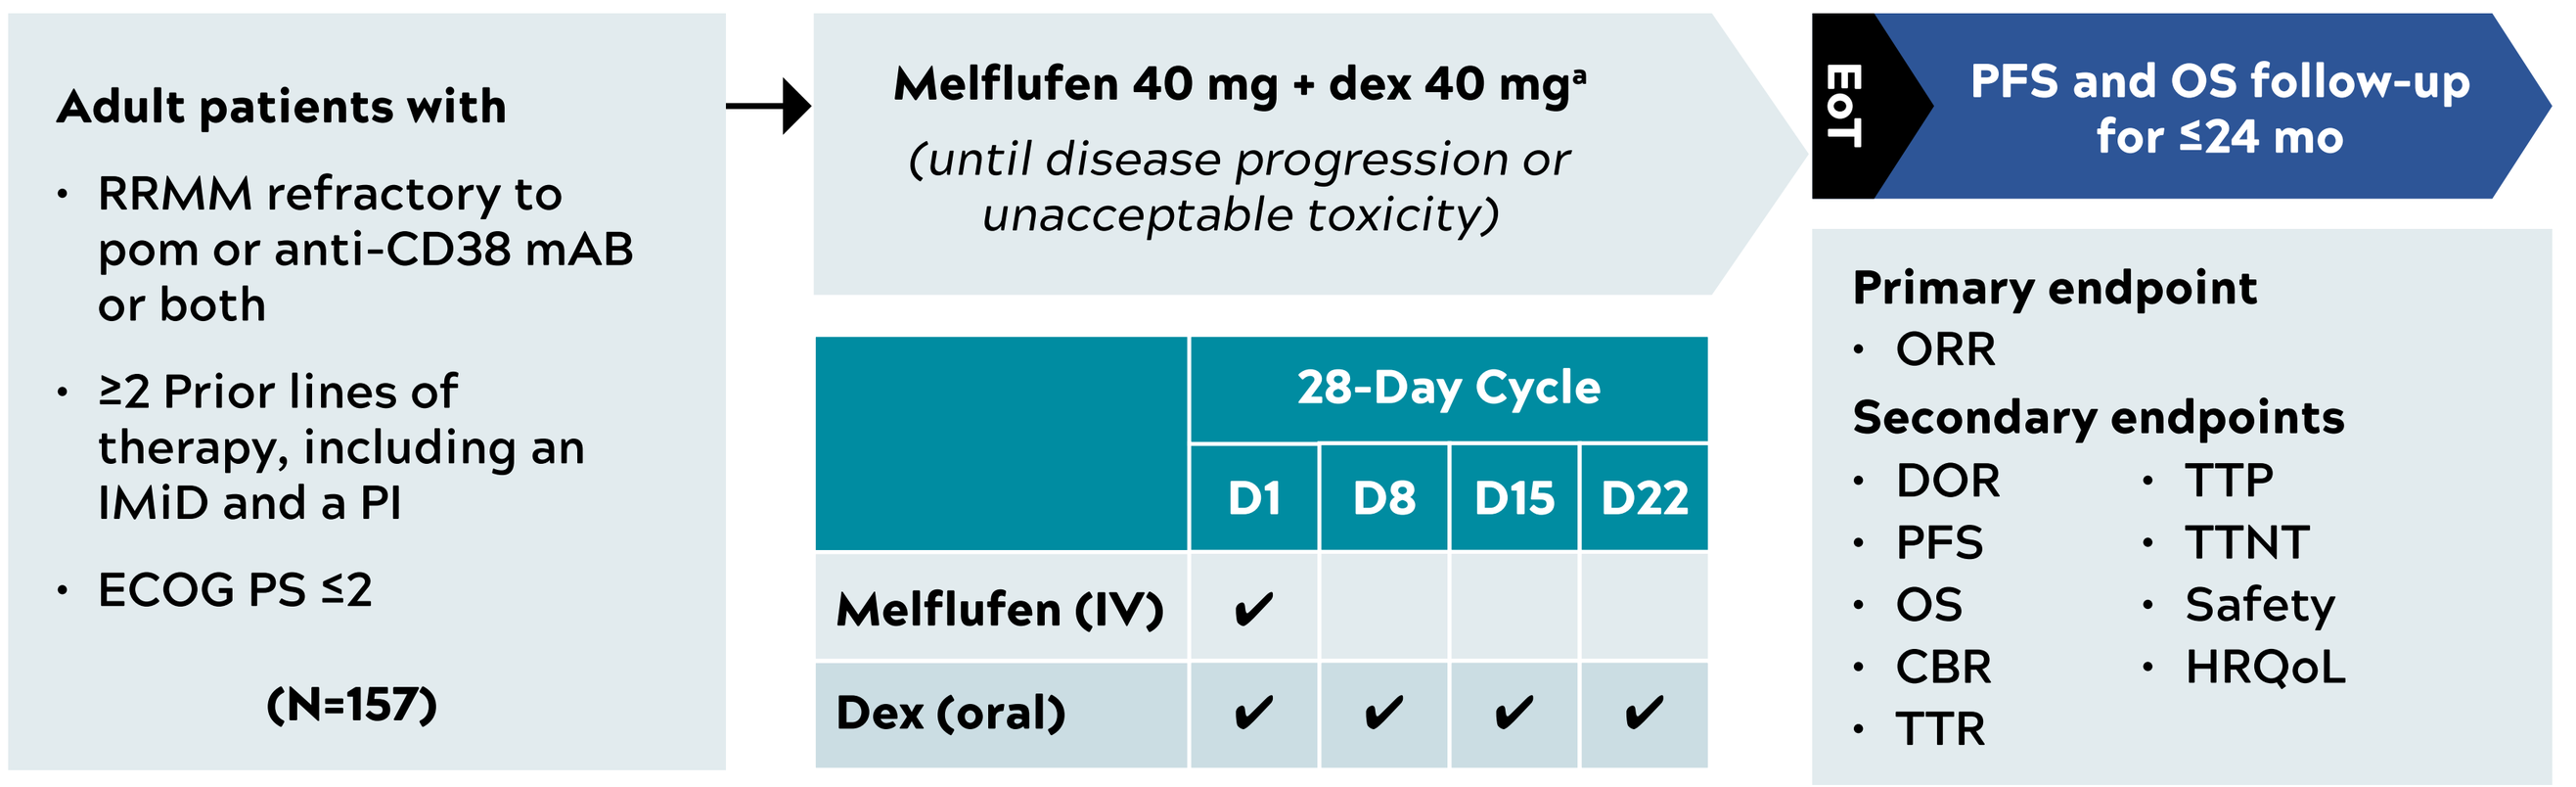

Adult patients with
• RRMM refractory to pom or anti-CD38 mAB or both
• ≥2 Prior lines of therapy, including an IMiD and a PI
• ECOG PS ≤2
(N=157)
Melflufen 40 mg + dex 40 mg[a]
(until disease progression or unacceptable toxicity)
28-Day Cycle
D1 D8 D15 D22
Melflufen (IV) ✔
Dex (oral) ✔ ✔ ✔ ✔
EoT
PFS and OS follow-up for ≤24 mo
Primary endpoint
• ORR
Secondary endpoints
• DOR • TTP
• PFS • TTNT
• OS • Safety
• CBR • HRQoL
• TTR

[a]Patients aged ≥75 years received dex 20 mg.
CBR, clinical benefit rate; dex, dexamethasone; DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group performance status; EoT, end of treatment; HRQoL, health-related quality of life; IMiD, immunomodulatory agent; IV, intravenous; mAb, monoclonal antibody; ORR, overall response rate; OS, overall survival; PFS, progression-free survival; PI, proteasome inhibitor; RRMM, relapsed/refractory multiple myeloma; TTNT, time to next treatment; TTP, time to progression; TTR, time to response.

RESULTS

- In total, 157 patients were enrolled and received ≥1 dose of study medication (data cutoff date, Jan 14, 2020); 131 patients (83%) had discontinued therapy and 26 patients (17%) remained on therapy
 - The most common primary reasons for treatment discontinuation were disease progression (56%) and AEs (17%)
- The study is fully recruited with median follow-up time of 14 months
 - The last patient enrolled into the study began treatment ≥3 months prior to the data cutoff date
- Baseline patient demographics and characteristics are shown in **Table 1**

Table 1. Baseline Patient Characteristics

Characteristic	ITT (N=157)	Triple-Class–Refractory (n=119)	EMD (n=55)
Median age (range), years	65 (35-86)	65 (35-86)	64 (43-82)
Male sex, n (%)	89 (57)	70 (59)	31 (56)
High-risk cytogenetics, n (%)[a]	59 (38)	41 (34)	19 (35)
ISS stage (I/II/III) at study entry, %[b]	40/31/25	34/30/30	36/25/33
EMD at study entry, n (%)[c]	55 (35)	50 (42)	55 (100)
Median no. of prior lines of therapy (range)	5 (2-12)	5 (2-12)	5 (2-12)
Triple-class–refractory, n (%)[d]	119 (76)	119 (100)	50 (91)
Refractory to ≥1 anti-CD38 mAb	125 (80)	119 (100)	50 (91)
Refractory to prior alkylator therapy[e]	92 (59)	76 (64)	33 (60)

[a] High-risk cytogenetics at study entry was based on fluorescence in situ hybridization defined as t(4;14), del(17/17p), and t(14;16) per Sonneveld P, et al[12]; 31 patients (20%) had unknown cytogenetics. Cytogenetic assessments were not centralized.
[b] At study entry, 6 patients in the ITT population had unknown or missing ISS stage.
[c] EMD was defined as a multiple myeloma disease originating either in, but extending beyond, the cortical bone or as a separate soft tissue mass.
[d] Defined as refractory to or intolerant of ≥1 proteasome inhibitor, ≥1 immunomodulatory drug, and ≥1 anti-CD38 mAb.
[e] Including 21 patients (13%) refractory to prior melphalan in the ITT population.
EMD, extramedullary disease; ISS, International Staging System; ITT, intention-to-treat; mAb, monoclonal antibody.

- The ORR was 29% (95% CI, 22-37) in the ITT population, 26% (95% CI, 18-35) in the triple-class–refractory population, and 24% (95% CI, 13-37) in the EMD subgroup (**Figure 3**), and were consistent with the findings of the independent review committee (IRC)

Figure 3. Best Overall Confirmed Response in the ITT and Triple-Class–Refractory Populations, and in Patients with EMD

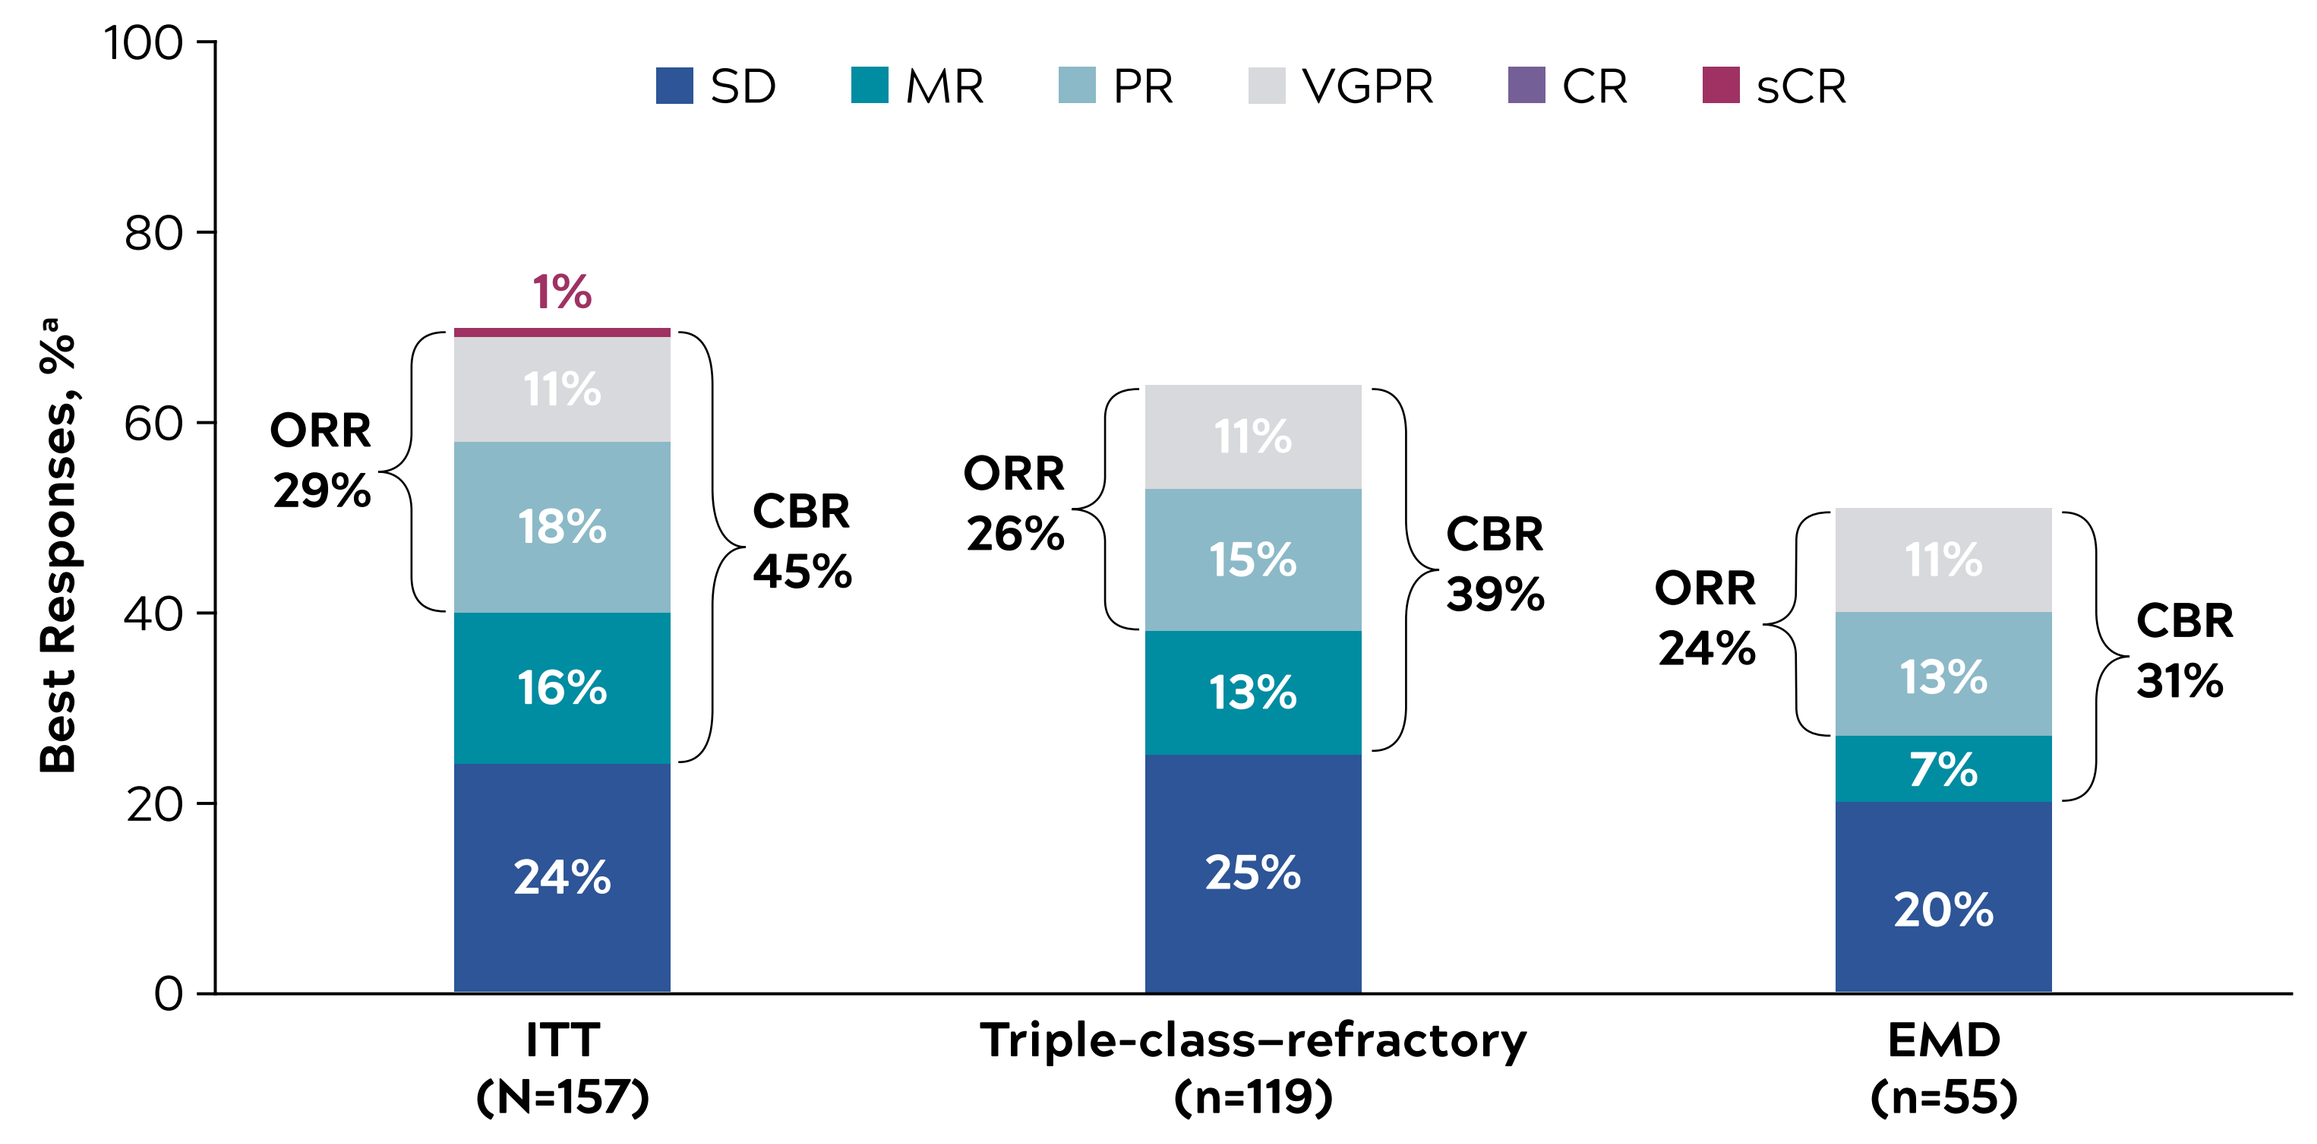

SD
MR
PR
VGPR
CR
sCR
Best Responses, %[a]
100
80
60
40
20
0
ITT (N=157)
1%
11%
18%
16%
24%
ORR 29%
CBR 45%
Triple-class–refractory (n=119)
11%
15%
13%
25%
ORR 26%
CBR 39%
EMD (n=55)
11%
13%
7%
20%
ORR 24%
CBR 31%

[a] Investigator-assessed best overall response per International Myeloma Working Group uniform criteria.[13]
CBR, clinical benefit rate; CR, complete response; EMD, extramedullary disease; ITT, intention-to-treat; MR, minor response; ORR, overall response rate; PR, partial response; sCR, stringent complete response; SD, stable disease; VGPR, very good partial response.

- Median DOR was 5.5 months (95% CI, 3.9-7.6) in the ITT population, 4.4 months (95% CI, 3.4-7.6) in the triple-class–refractory population, and 5.5 months (95% CI, 1.8-not evaluable) in the EMD subgroup
- Median time to best response was 1.9 months in the ITT population, and responses deepened with longer treatment duration (**Figure 4**)
 - In some patients, response was extended beyond last dose of melflufen
- Among patients with a response (≥PR), median PFS was 8.5 months (95% CI, 5.4-13.4) in the ITT population, 8.5 months (95% CI, 5.3-13.4) in the triple-class–refractory population, and 17.3 months (95% CI, 5.3-not evaluable) in the EMD subgroup

Figure 4. Patients in the ITT Population Who Achieved a Partial Response or Better

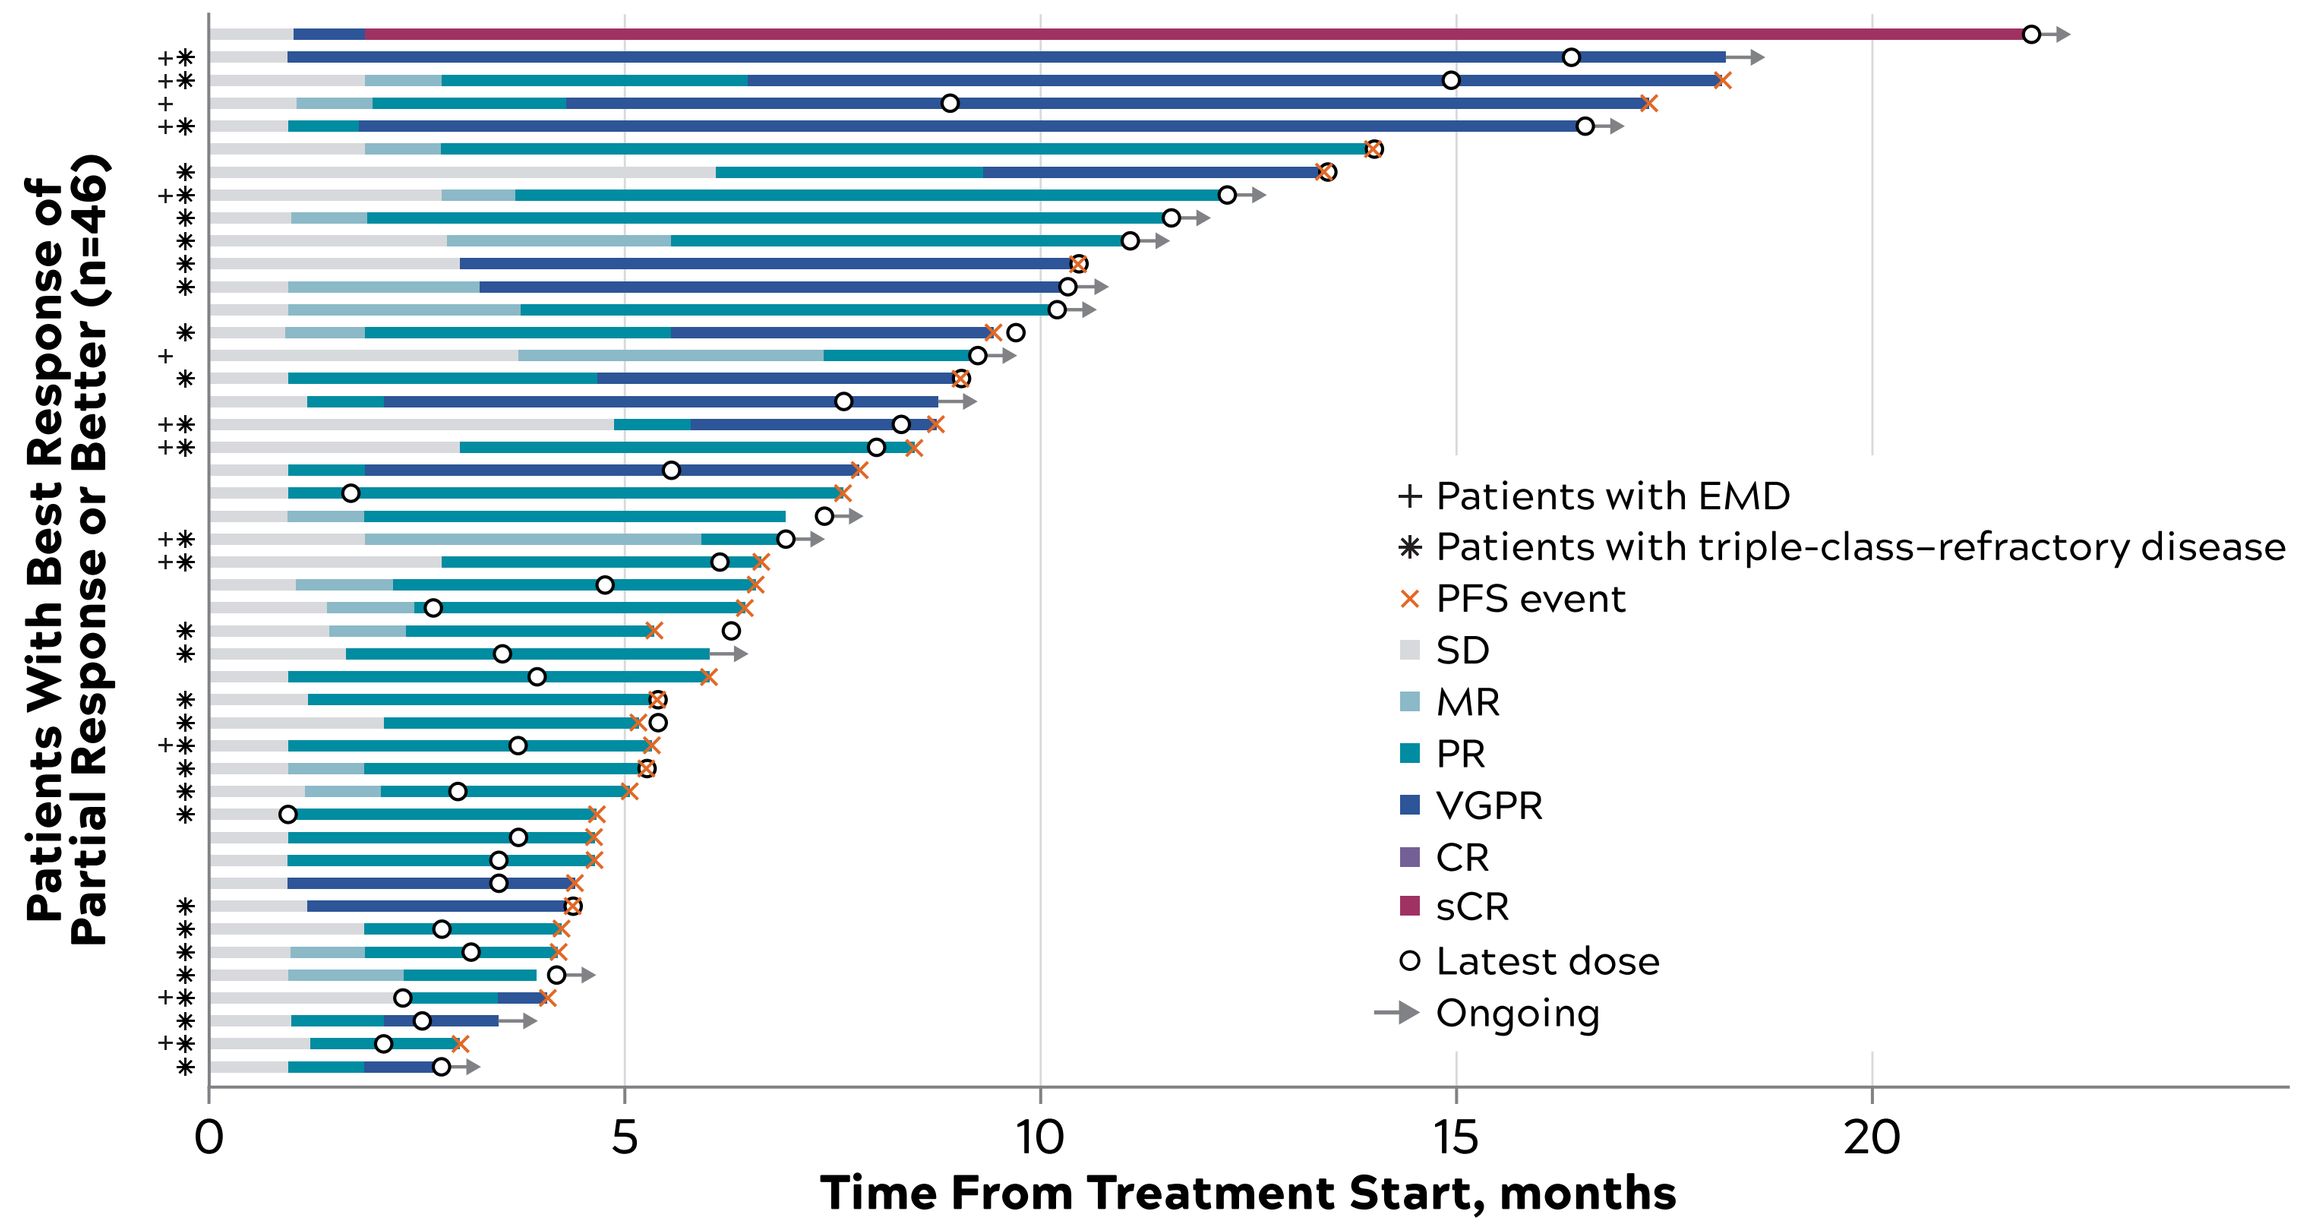

Patients With Best Response of Partial Response or Better (n=46)
Time From Treatment Start, months
0
5
10
15
20
+ Patients with EMD
✱ Patients with triple-class–refractory disease
× PFS event
SD
MR
PR
VGPR
CR
sCR
Latest dose
Ongoing

[a] Investigator-assessed best overall response per International Myeloma Working Group uniform criteria.[13]
CR, complete response; EMD, extramedullary disease; ITT, intention-to-treat; MR, minimal response; PFS, progression-free survival; PR, partial response; sCR, stringent complete response; SD, stable disease; VGPR, very good partial response.

- Median PFS was 4.2 months (95% CI, 3.4-4.9) in the ITT population, 3.9 months (95% CI, 3.0-4.6) in the triple-class–refractory population, and 2.9 months (95% CI, 2.0-3.8) in patients with EMD (**Figure 5**)

Figure 5. PFS in the ITT and Triple-Class–Refractory Populations, and in Patients with EMD

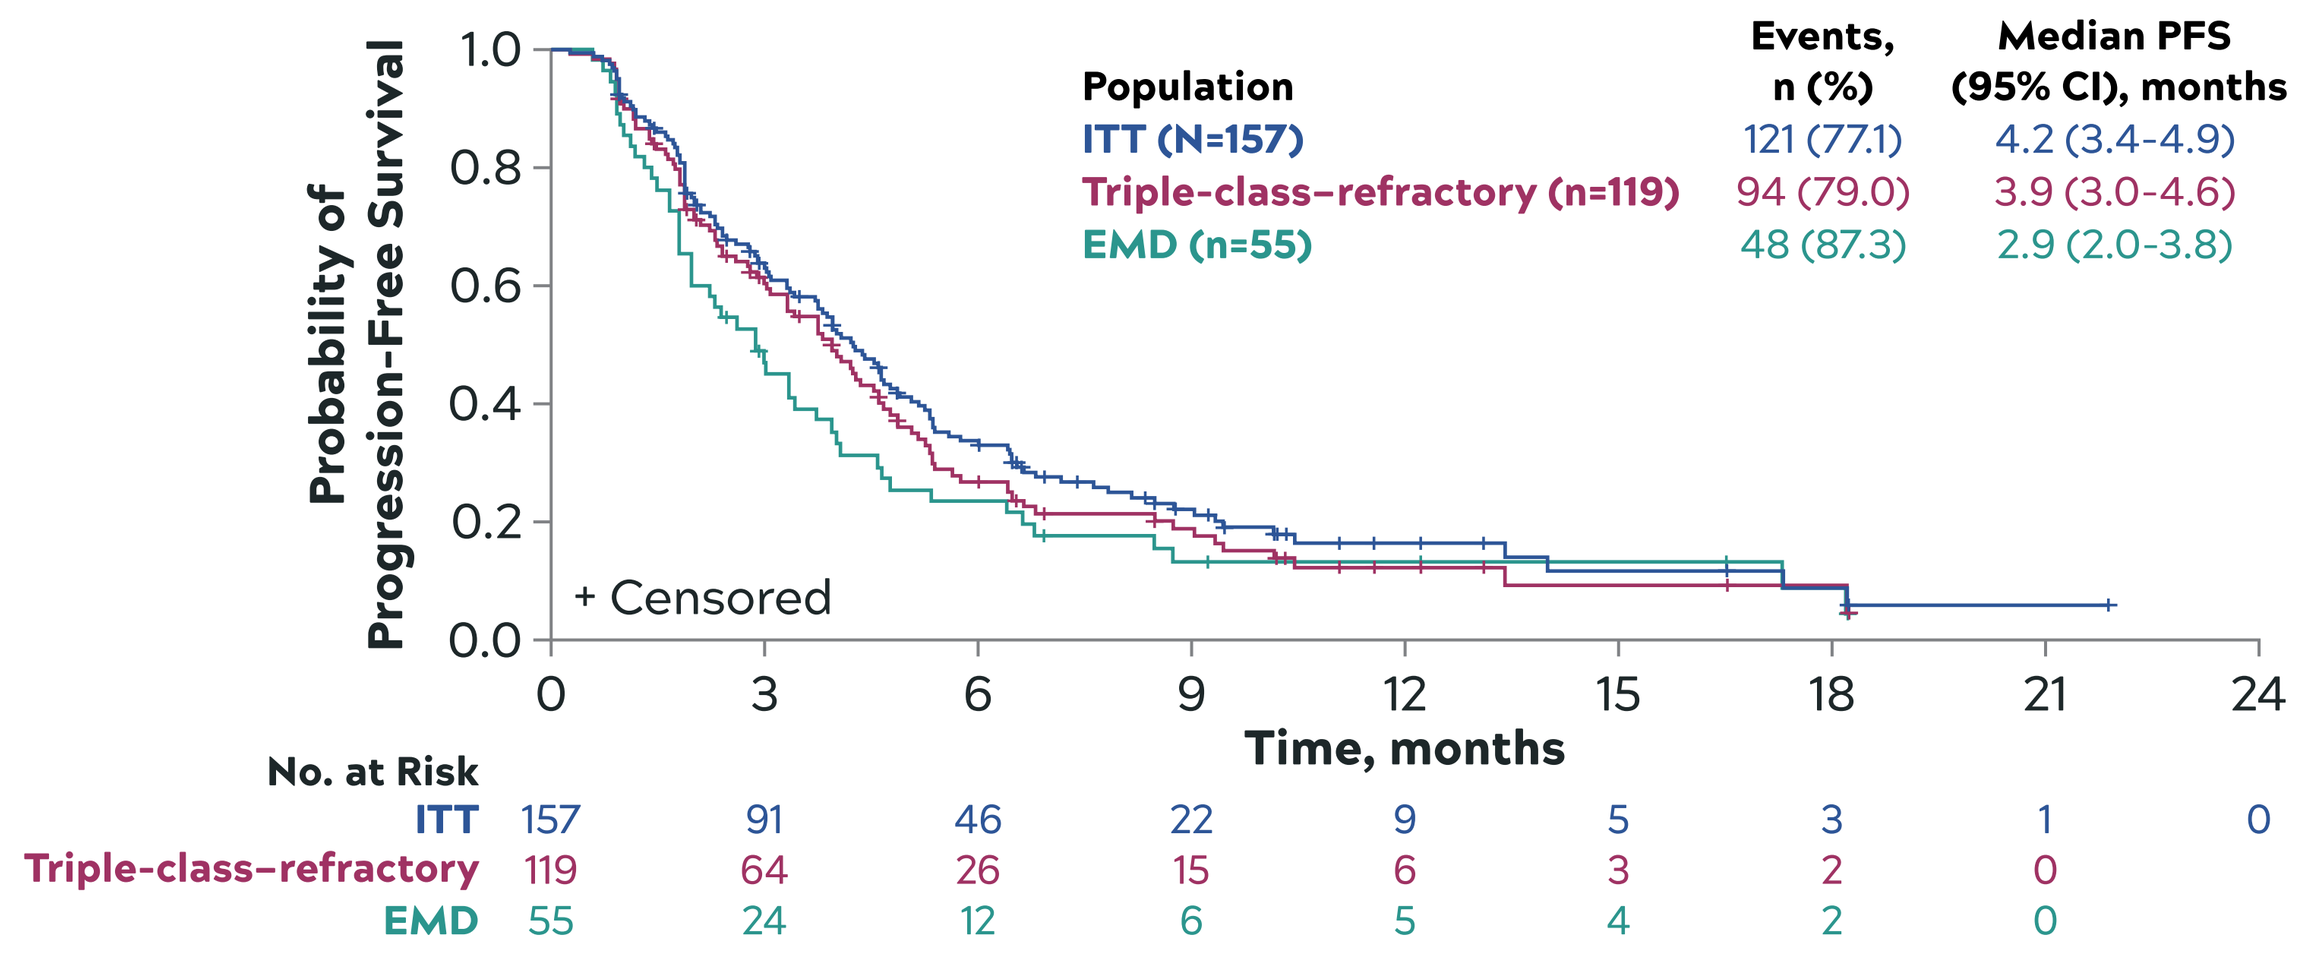

Probability of Progression-Free Survival
1.0
0.8
0.6
0.4
0.2
0.0
+ Censored
Time, months
0 3 6 9 12 15 18 21 24
Population | Events, n (%) | Median PFS (95% CI), months
ITT (N=157) | 121 (77.1) | 4.2 (3.4-4.9)
Triple-class–refractory (n=119) | 94 (79.0) | 3.9 (3.0-4.6)
EMD (n=55) | 48 (87.3) | 2.9 (2.0-3.8)
No. at Risk
ITT 157 91 46 22 9 5 3 1 0
Triple-class–refractory 119 64 26 15 6 3 2 0
EMD 55 24 12 6 5 4 2 0

EMD, extramedullary disease; ITT, intention-to-treat; PFS, progression-free survival.

- Median OS was 11.6 months (95% CI, 9.3-15.4) in the ITT population, 11.2 months (95% CI, 7.7-13.2) in the triple-class–refractory population, and 6.5 months (95% CI, 5.1-9.7) in patients with EMD (**Figure 6**)

Figure 6. OS in the ITT and Triple-Class–Refractory Populations, and in Patients with EMD

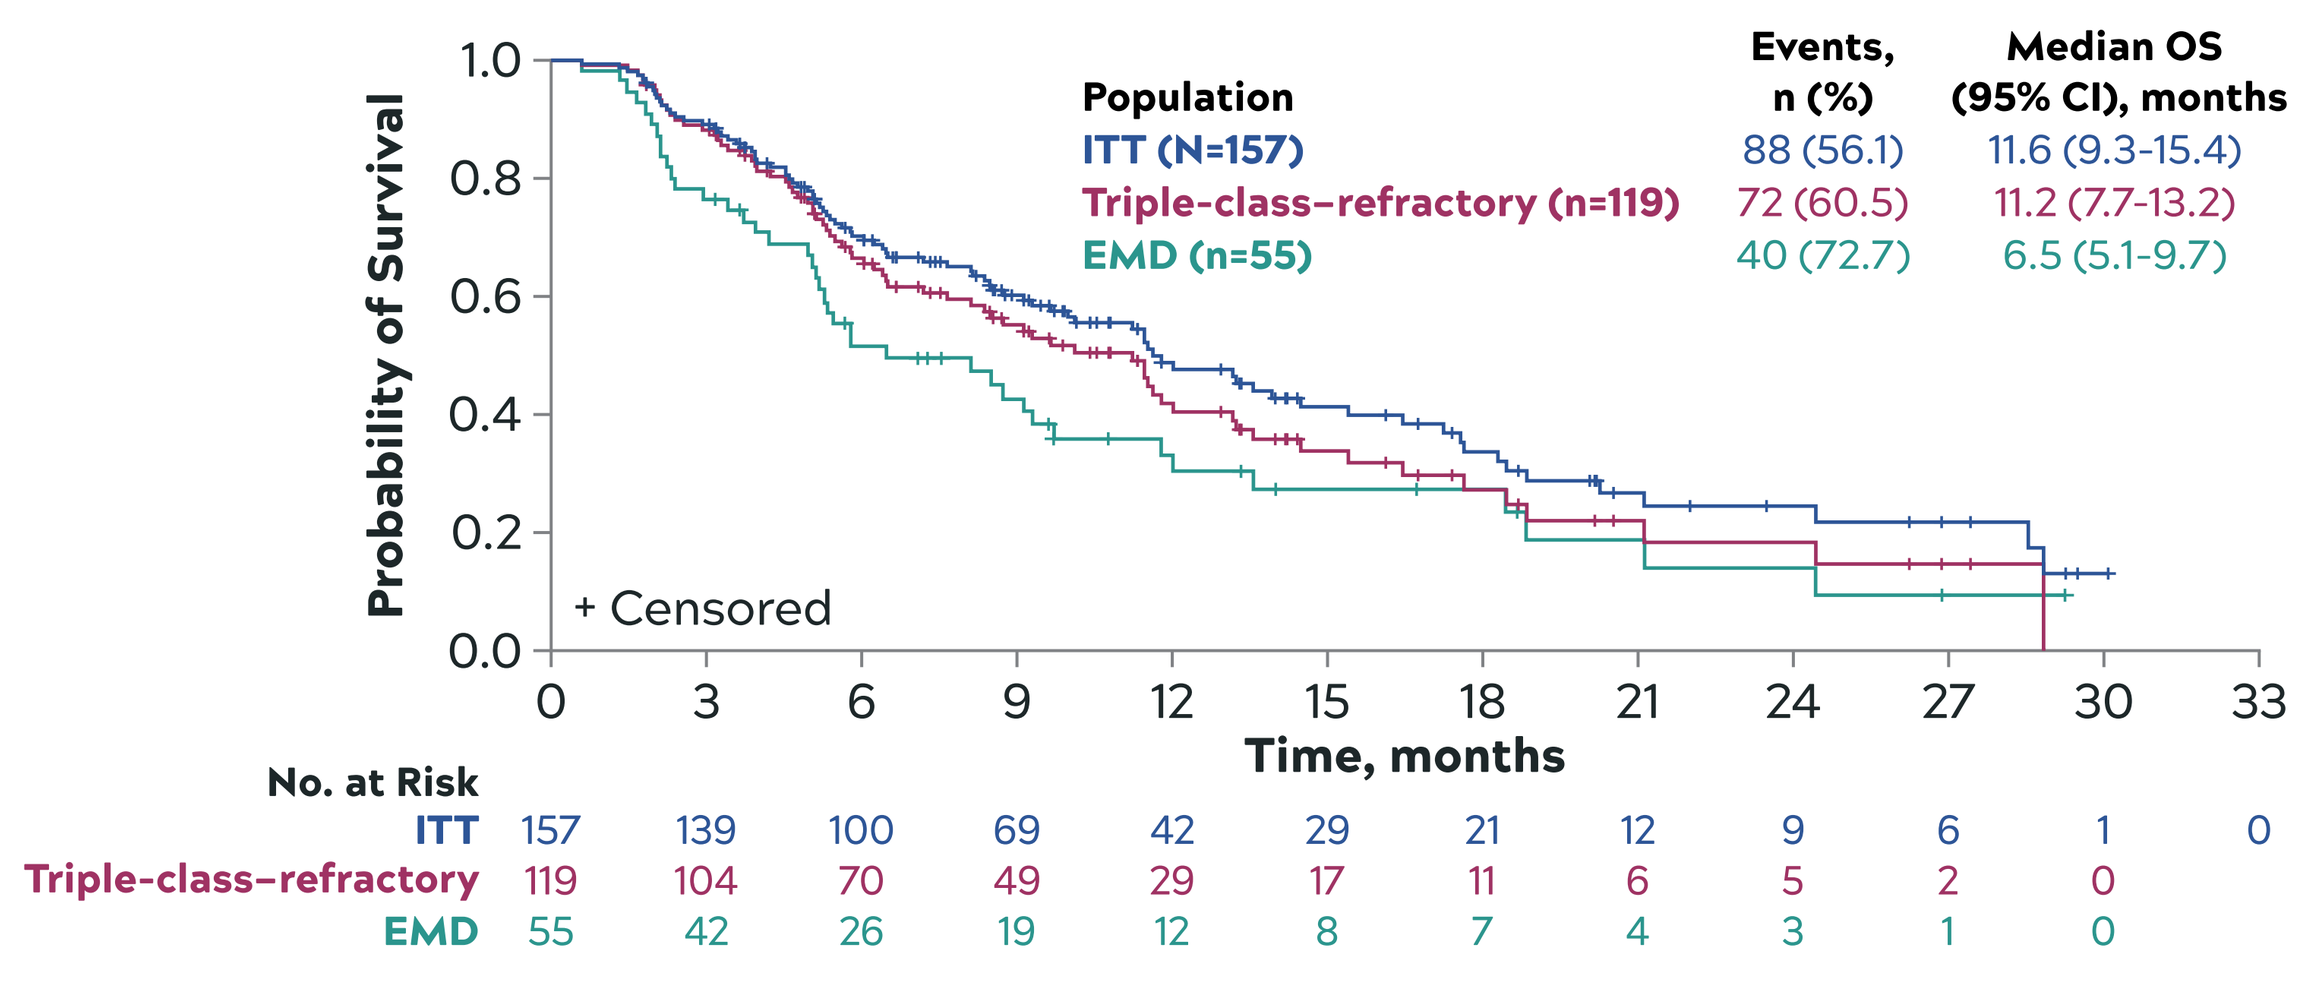

Probability of Survival
1.0
0.8
0.6
0.4
0.2
0.0
+ Censored
Time, months
0 3 6 9 12 15 18 21 24 27 30 33
Population | Events, n (%) | Median OS (95% CI), months
ITT (N=157) | 88 (56.1) | 11.6 (9.3-15.4)
Triple-class–refractory (n=119) | 72 (60.5) | 11.2 (7.7-13.2)
EMD (n=55) | 40 (72.7) | 6.5 (5.1-9.7)
No. at Risk
ITT 157 139 100 69 42 29 21 12 9 6 1 0
Triple-class–refractory 119 104 70 49 29 17 11 6 5 2 0
EMD 55 42 26 19 12 8 7 4 3 1 0

EMD, extramedullary disease; ITT, intention-to-treat; OS, overall survival.

- Any-grade and grade 3/4 AEs occurred in 100% and 94% of patients, respectively
- The most frequent AEs overall (occurring in ≥ 10% of patients in the ITT population) and by maximum grade are shown in **Table 2**
 - The most common grade 3/4 AEs were neutropenia (79%), thrombocytopenia (76%), and anemia (43%)
 - The most common nonhematologic grade 3/4 AE was pneumonia (10%)

Table 2. AEs Occurring in ≥10% of Patients in the ITT Population

	AEs (N=157), n (%)				
	Any-Grade	Grade 1	Grade 2	Grade 3	Grade 4
Any AE[a]	**157 (100)**	**0**	**7 (4)**	**40 (25)**	**100 (64)**
Hematologic[b]					
Neutropenia	129 (82)	1 (<1)	4 (3)	50 (32)	74 (47)
Thrombocytopenia	128 (82)	5 (3)	3 (2)	40 (25)	80 (51)
Anemia	111 (71)	3 (2)	41 (26)	66 (42)	1 (<1)
Nonhematologic					
Nausea	50 (32)	31 (20)	18 (11)	1 (<1)	0
Fatigue	46 (29)	17 (11)	25 (16)	4 (3)	0
Asthenia	42 (27)	13 (8)	23 (15)	5 (3)	1 (<1)
Diarrhea	42 (27)	24 (15)	18 (11)	0	0
Pyrexia	38 (24)	24 (15)	11 (7)	3 (2)	0
Cough	26 (17)	16 (10)	10 (6)	0	0
Upper respiratory tract infection	25 (16)	3 (2)	19 (12)	3 (2)	0
Constipation	23 (15)	18 (11)	4 (3)	1 (<1)	0
Decreased appetite	22 (14)	10 (6)	11 (7)	1 (<1)	0
Hypokalemia	22 (14)	14 (9)	6 (4)	2 (1)	0
Peripheral edema	22 (14)	15 (10)	5 (3)	2 (1)	0
Headache	21 (13)	13 (8)	8 (5)	0	0
Vomiting	21 (13)	12 (8)	9 (6)	0	0
Bone pain	20 (13)	9 (6)	8 (5)	3 (2)	0
Pain in extremity	20 (13)	7 (4)	10 (6)	3 (2)	0
Pneumonia	20 (13)[c]	0	3 (2)	14 (9)	2 (1)
Back pain	19 (12)	9 (6)	9 (6)	1 (<1)	0
Insomnia	18 (11)	14 (9)	3 (2)	1 (<1)	0
Dizziness	17 (11)	14 (9)	3 (2)	0	0
Dyspnea	17 (11)	9 (6)	6 (4)	2 (1)	0
Arthralgia	16 (10)	11 (7)	5 (3)	0	0
Exertional dyspnea	16 (10)	13 (8)	3 (2)	0	0
Hypocalcemia	16 (10)	9 (6)	6 (4)	1 (<1)	0

[a] Treatment-emergent adverse events by maximum severity. Adverse events are coded to preferred term using MedDRA, version 19.1.
[b] Hematologic AEs of special interest (neutropenia, thrombocytopenia, and anemia) were categorized by standardized MedDRA query.
[c] Includes one grade 5 event.
AE, adverse event; ITT, intention-to-treat.

- The clinical implications of thrombocytopenia and neutropenia were limited
 - Grade 3/4 thrombocytopenia with concurrent grade 3/4 bleeding events occurred in 4 patients (3%; 1 was grade 4)
 - Grade 3/4 neutropenia with concurrent grade 3/4 infections occurred in 18 patients (11%; 1 was grade 4)
- AEs led to melflufen dose reductions in 42 patients (27%) and dose delays in 95 patients (61%)
 - The most common AE leading to melflufen dose reductions was thrombocytopenia (14%)
- Serious AEs occurred in 49% of patients overall; most commonly pneumonia (9%) and febrile neutropenia (5%)
- Fatal AEs occurred in 10 patients (6%); none were considered related to melflufen

CONCLUSIONS

- Melflufen plus dex demonstrated clinically meaningful efficacy and a manageable safety profile in patients with heavily pretreated RRMM, including those with triple-class–refractory disease and EMD
- These results are consistent with previously reported data,[11] but in an RRMM population that is more reflective of current treatment practices in earlier lines of therapy[14]
- Responses were durable and deepened with prolonged melflufen plus dex treatment, suggesting that patients should remain on treatment for as long as possible
- The safety profile of melflufen plus dex was consistent with previous reports, and no new safety concerns were identified[11]
 - The safety profile of melflufen plus dex consisted primarily of hematologic adverse events that were clinically manageable with dose modifications and supportive care
 - Gastrointestinal events were mostly grade 1/2 and did not result in treatment discontinuation
- Melflufen, with its novel mechanism of action, in combination with dex has the potential to provide a needed therapeutic option to patients with poor-risk and heavily pretreated RRMM. Results from HORIZON will support a submission to the US Food and Drug Administration for the accelerated approval of melflufen in this hard to treat patient population
- The efficacy and safety of melflufen plus dex are being further evaluated in OCEAN (OP-103), a randomized, head-to-head, superiority, open-label, global, phase 3 study of melflufen plus dex vs pom plus dex in patients with RRMM refractory to lenalidomide (NCT03151811)

REFERENCES

1. Kumar SK, et al. *Leukemia*. 2017;31:2443.
2. Gandhi UH, et al. *Leukemia*. 2019;33:2266.
3. Pawlyn C, et al. EHA 2019. Abstract S873.
4. Moreau P, et al. *Blood Cancer J*. 2019;9:38.
5. Cejalvo MJ, et al. *Expert Rev Hematol*. 2017;10:383-392.
6. Chauhan D, et al. *Clin Cancer Res*. 2013;19(11):3019-3031.
7. Wickström M, et al. *Invest New Drugs*. 2008;26(3):195-204.
8. Ray A, et al. *Br J Haematol*. 2016;174(3):397-409.
9. Strese S, et al. *Biochem Pharmacol*. 2013;86(7):888-895.
10. Wickström M, et al. *Oncotarget*. 2017;8(39):66641-66655.
11. Richardson PG, et al. *Lancet Haematol*. 2020;7:e395-e407.
12. Sonneveld P, et al. *Blood*. 2016;127:2955-2962.
13. Rajkumar SV, et al. *Blood*. 2011;117:4691-4695.
14. Richardson PG, et al. *Blood Cancer J*. 2018;8:109.

ACKNOWLEDGMENTS

The authors would like to thank the patients who volunteered to participate in the study, the staff and the study sites who cared for them, and Precision for Medicine, the CRO involved in data gathering and analyses.

Medical writing support was provided by Katherine Mills-Lujan, PhD, CMPP, of Team 9 Science with funding from Oncopeptides..

DISCLOSURES

PGR received grant support from Bristol-Myers Squibb, grant support and honoraria from Celgene, Karyopharm, Oncopeptides, and Takeda, and honoraria from Janssen, Sanofi, and SecuraBio. **AO** reports consultancy for Amgen, Celgene-Bristol-Myers Squibb, and Janssen. **AL** reports honoraria and personal fees from Bristol-Myers Squibb, Celgene, Janssen, and Takeda, and personal fees from Amgen and GlaxoSmithKline. **JB** reports honoraria from Amgen, Celgene, Janssen, Oncopeptides, and Takeda. **MC** received personal fees from AbbVie, Amgen, Bristol- Myers Squibb, Celgene, GlaxoSmithKline, Janssen, Sanofi, and Takeda. **PRO** reports honoraria from AbbVie, Amgen, Celgene-Bristol-Myers Squibb, GlaxoSmithKline, Janssen, Kite, Oncopeptides, and Sanofi. **ON** reports personal fees from Amgen, Celgene, Janssen, Sanofi, and Takeda. **HH** received grant support from Celgene and Janssen, and personal fees from Novartis. **CT** reports personal fees from Oncopeptides during the conduct of the study. **AA** received honoraria from Amgen, Bristol-Myers Squibb, Celgene, GlaxoSmithKline, Janssen, Oncopeptides, Sanofi, and Takeda. **AP** reports honoraria from Amgen and Celgene and consultancy/advisory fees from AbbVie, Cellectar, Janssen, Karyopharm, and Takeda. **CM** received honoraria from Amgen, Celgene, Incyte, Janssen, Karyopharm, Kite, Takeda, and Verastem, and consulting fees from Bayer. **XL, MN, JWH, AM, AR,** and **NP** have nothing to disclose. **JH** and **CB** are consultants of and owns stock and stock options in Oncopeptides. **ST** and **JO** are employees of and own stock and stock options in Oncopeptides. **MVM** received personal fees from AbbVie, Adaptive, Amgen, Celgene, GlaxoSmithKline, Janssen, Roche, Seattle Genetics, and Takeda.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from EHA® and the author of this poster.

Presented at the 25th Congress of the European Hematology Association (EHA25 Virtual), June 11-21, 2020

oncopeptides