Melflufen therapy for Relapsed Refractory Multiple Myeloma (RRMM) patients Refractory to Daratumumab and/or Pomalidomide; an early report on efficacy

HORIZON

María-Victoria Mateos, MD[1], Albert Oriol Rocafiguera, MD[2], Paula Rodríguez Otero, MD[3], Joan Bladé, MD[4], Alessandra Larocca, MD[5], Jan S. Moreb, MD[6], Michele Cavo, MD[7], Amitabha Mazumder, MD[8], Adrián Alegre, MD[9], Hani Hassoun, MD[10], Christopher Maisel, MD[11], Agne Paner, MD[12], Nashat Gabrail, MD[13], Kathleen Halka, MD[14], Jeffrey Zonder, MD[15], Enrique Ocio, MD[1], Catriona Byrne, RN[16], Johan Harmenberg, MD[16], Jakob Lindberg, MSc[16], Eva Nordström, MSc[16], Sara Thuresson, MSc[16] and Paul G. Richardson, MD[17]

[1]Hospital Clínico Universitario de Salamanca, Salamanca, Spain; [2]ICO Badalona - Hospital Germans Trias i Pujol, Badalona, Spain; [3]Clínica Universidad de Navarra, Pamplona, Spain; [4]Hospital Clínica de Barcelona, Servicio de Onco-Hematología, Barcelona, Spain; [5]A.O.U. Città della Salute e della Scienza di Torino – S.C. Ematologia U., Torino, Italy; [6]UF Health Shands Cancer Hospital, Gainesville, FL, USA; [7]Policlinico S.Orsola Malphigi, Bologna, Italy; [8]Hudson Valley Hematology Oncology Associates RLLP, Hawthorne, NY, USA; [9]Hospital Universitario La Princesa, Madrid, Spain; [10]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [11]Baylor Scott & White Charles A Sammons Cancer Center, Dallas, TX, USA; [12]Rush University Medical Center, Chicago, IL, USA; [13]Gabrail Cancer Center Research, Canton, OH, USA; [14]Scott & White Memorial Hospital and Clinic, Temple, TX, USA; [15]Karmanos Cancer Institute, Detroit, MI, USA; [16]Oncopeptides AB, Stockholm, Sweden; [17]Dana-Farber Cancer Institute, Harvard Medical School, Boston, MA, USA

BACKGROUND

Melflufen is a next generation alkylator, belonging to the novel class of Peptidase Enhanced Cytotoxics (PEnCs), designed for efficient targeting of tumor cells with a unique mechanism of action. Melflufen provides a peptidase enhanced therapy with an alkylating payload and triggers fast, robust and irreversible DNA damage. The lipophilicity of melflufen leads to rapid and extensive distribution into cells where it is readily metabolized by intracellular peptidases (often over-expressed in malignant cells) into hydrophilic alkylating metabolites leading to 50-fold enrichment of these metabolites in multiple myeloma (MM) cells. In addition, melflufen has potent anti-angiogenic properties.

Melflufen is a peptidase enhanced therapy with an alkylating payload

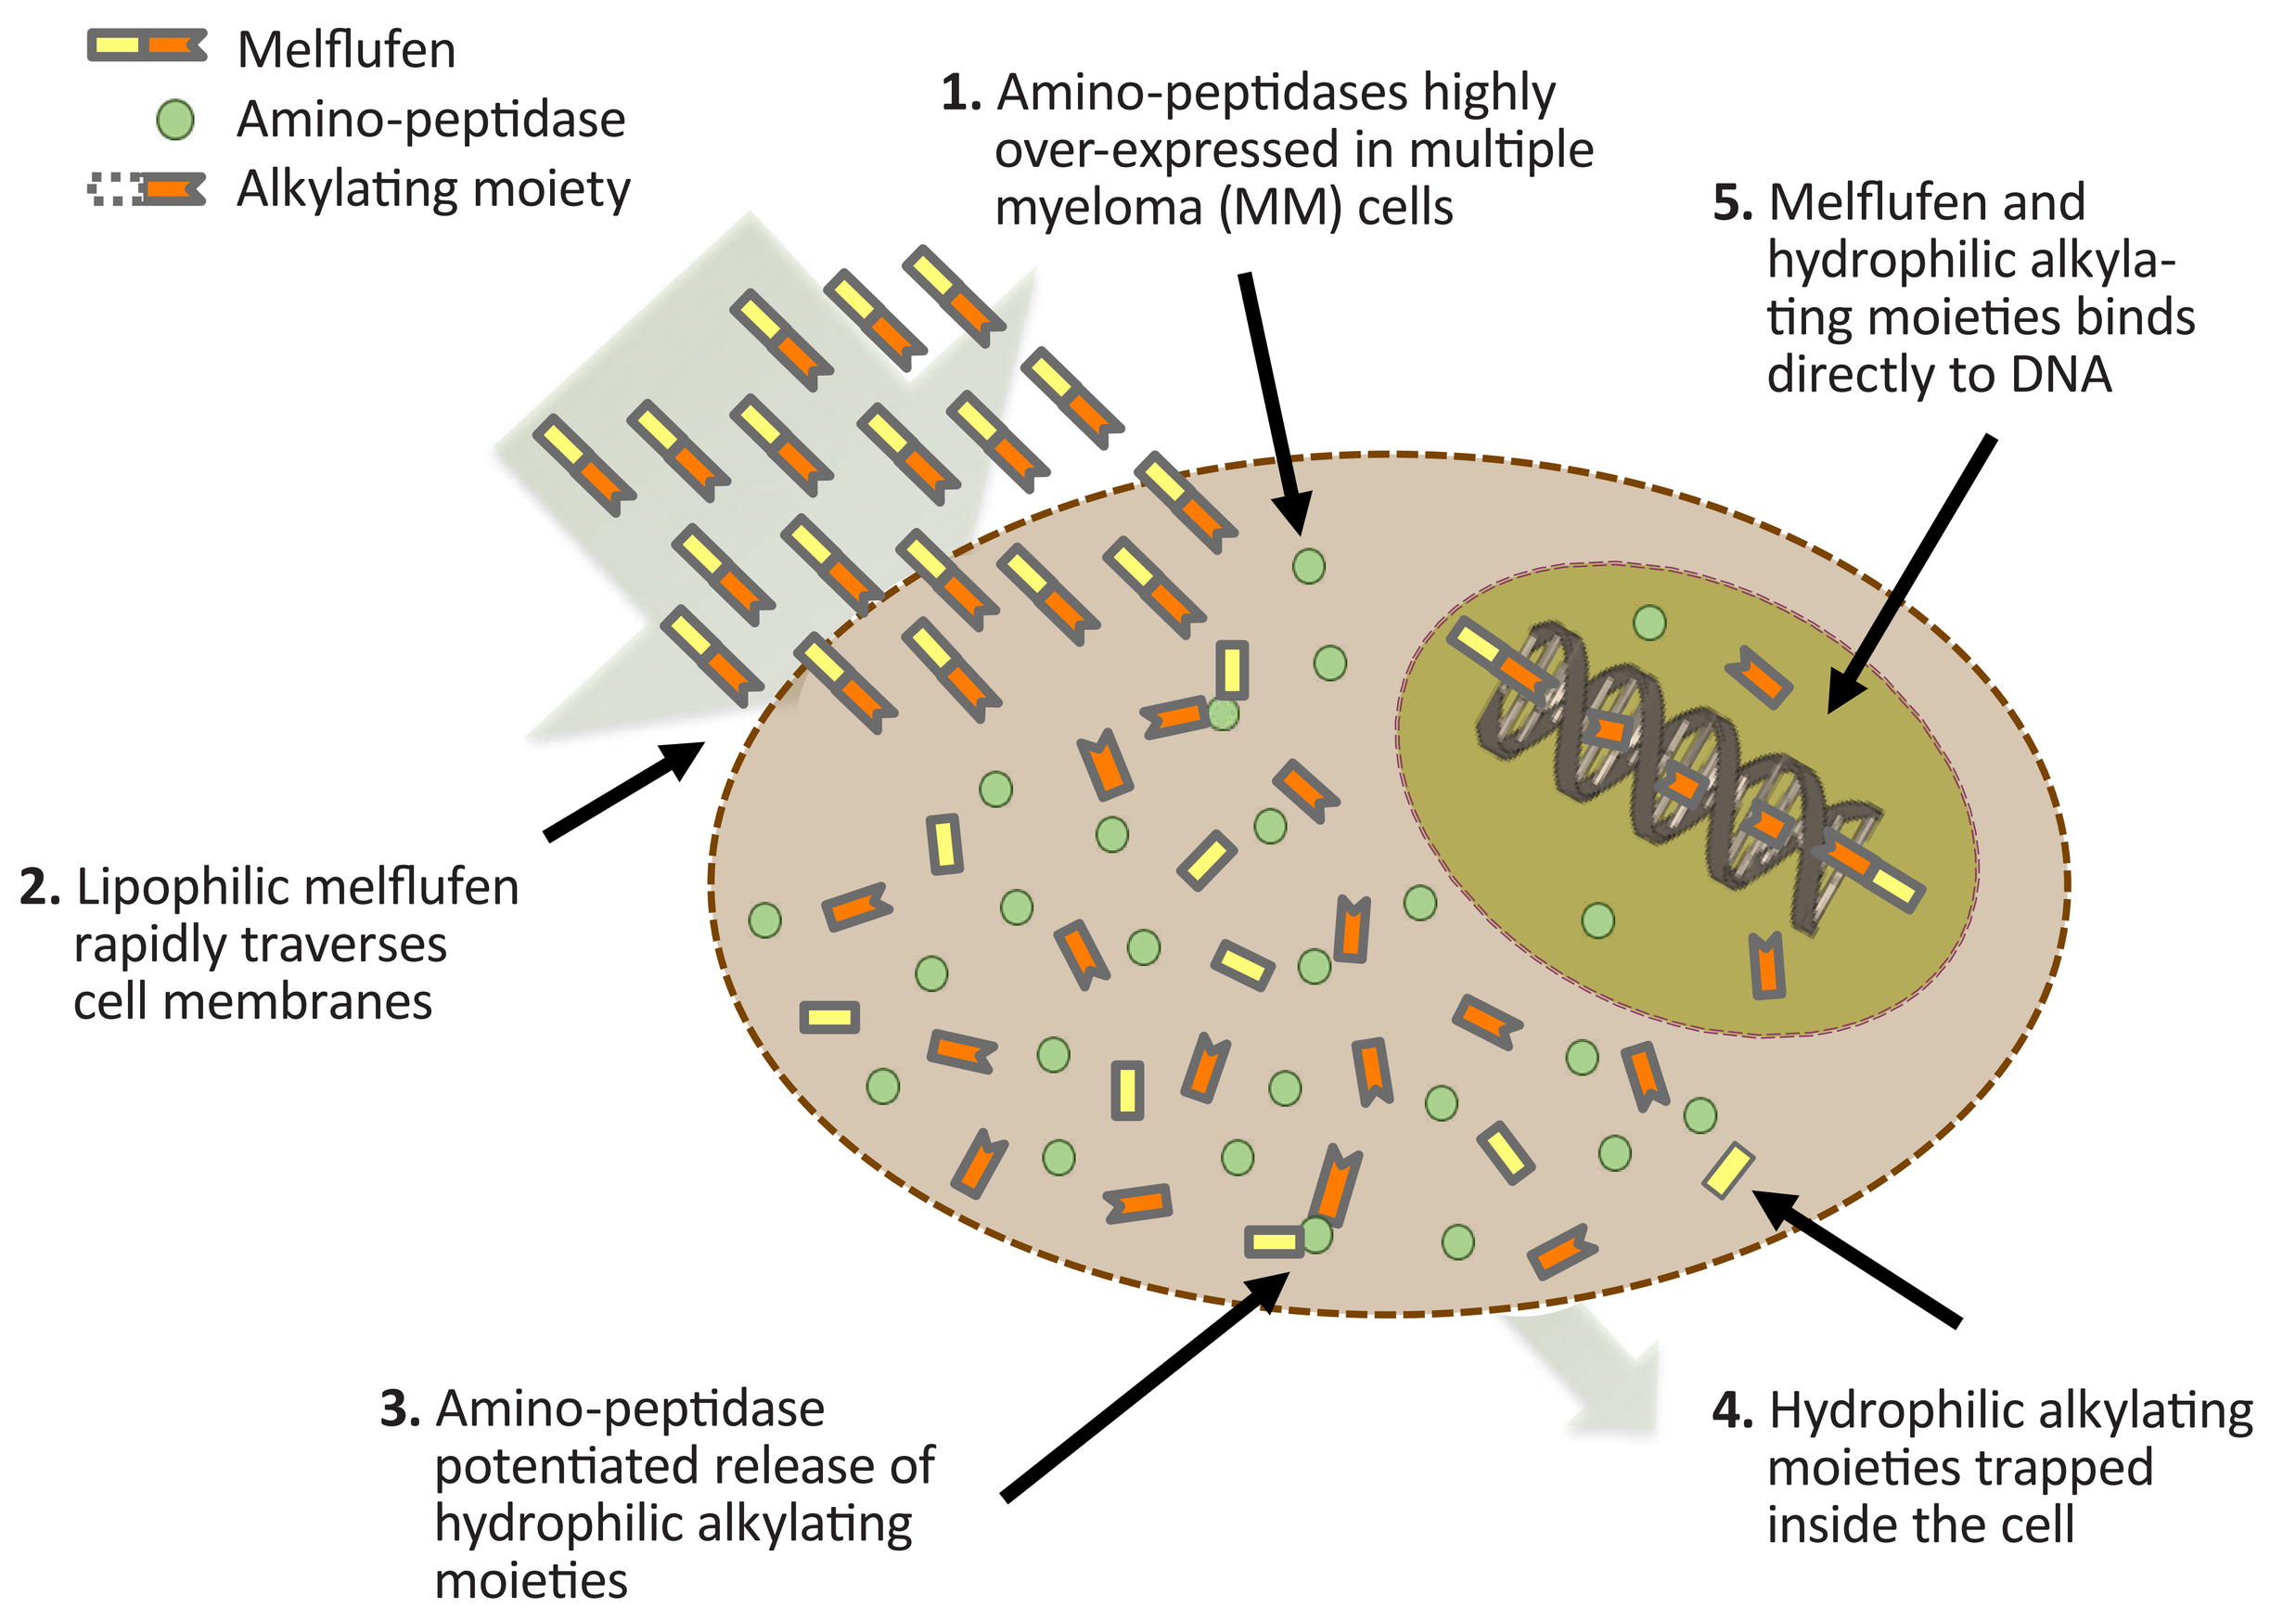
Melflufen
Amino-peptidase
Alkylating moiety
1. Amino-peptidases highly over-expressed in multiple myeloma (MM) cells
2. Lipophilic melflufen rapidly traverses cell membranes
3. Amino-peptidase potentiated release of hydrophilic alkylating moieties
4. Hydrophilic alkylating moieties trapped inside the cell
5. Melflufen and hydrophilic alkylating moieties binds directly to DNA

Peptidase enhanced activity in MM cells results in:

- Approx. 50-fold higher intra-cellular exposure in MM cells[1,5]
- Approx. 50-fold higher anti-MM potency[1,2,5]
- Alkylation of DNA with limited or no induction of DNA repair[3,5]
- Strong anti-angiogenic properties[1,4,5]
- Therapeutic index of 20x – 40x (MM cells compared with peripheral blood mononuclear cells)[1,5]

1. Chauhan et al. (2013) Clin Cancer Res 19(11): 3019-303. 2. Wickstrom et al. (2008) Invest New Drugs 26(3): 195-204.
3. Ray et al. (2016) Br J Hematol 174: 397-409. 4. Strese et al. (2013) Biochem Pharmacol 86: 888–895.
5. Wickström et al. (2017) Oncotarget E-pub June 08.

METHODS

Melflufen 40 mg is given i.v. on Day 1 of each 28-day cycle, with dexamethasone 40 mg weekly, in relapsed-refractory multiple myeloma (RRMM) patients refractory to pomalidomide and/or daratumumab with measurable disease and at least 2 prior lines of therapy including an IMiD and a PI (NCT02963493). Response is investigator assessed at each cycle by IMWG criteria. The primary objective is overall response rate (ORR). Patients receive treatment until there is documented disease progression or unacceptable toxicity.

ASH abstract no. 1841

BASELINE CHARACTERISTICS AND DISPOSITION

The study was initiated in Dec 2016. Thirty-eight patients were included at data cut-off 13 Nov 2017. The median time from initial diagnosis was 6.3 years (0.6–16). The median number of prior therapies was 6 (3–11). Thirty-two (86%) patients were double-refractory (IMiD + PI), 38 (100%) patients were refractory to pomalidomide or daratumumab and 23 (62%) were refractory to pomalidomide and daratumumab.

Table 1. Baseline characteristics (N=38)

CHARACTERISTICS	
Median age, years (range)	61.5 (41-87)
Median years since diagnosis, years (range)	6.3 (0.6–16)
Number of previous lines (range)	6 (3-11)
ISS at study entry, n (%)	
I	9 (24)
II	7 (18)
III	19 (50)
Unknown	3 (8)
ECOG performance status, n (%)	
0	10 (26)
1	23 (61)
2	5 (13)
High risk, cytogenetic risk factor by FISH*, n (%)	18 (47)

* [t(4;14), t(14;16), t(14;20), del(17/17p) or gain(1q)]

Table 2. Characteristics of prior lines of therapy (N=38)

CHARACTERISTICS	n (%)
Double-refractory (IMiD + PI)	32 (86)
Alkylator exposed	34 (89)
Alkylator refractory	21 (57)
Last line refractory (progressed while on therapy or within 60 days of last dose)*	36 (97)
Pomalidomide refractory	35 (96)
Daratumumab refractory	26 (68)
Pomalidomide or Daratumumab refractory	38 (100)
Pomalidomide and Daratumumab refractory	23 (62)

* 1 patient had incomplete data. 1 patient had PD 62 days after the last dose

Treatment is ongoing in 17 (45%) patients. Three (8%) patients discontinued treatment due to adverse events (AEs), 17 (45%) due to disease progression and 1 patient due to physician´s decision.

Table 3. Patient disposition (N=38)

	DISCONTINUED TREATMENT		
ON TREATMENT	AEs	PD	PHYSICIAN'S DECISION
17	3	17	1

RESULTS – EFFICACY

At the time of data cut-off, a total of 92 doses of melflufen had been given (1–8 cycles). Twenty-seven (71%) patients had completed at least two cycles of melflufen. Median number of cycles was 2 (1–8).

Patient responses were assessed by the investigators and the overall response rate (ORR) was 27% among 30 patients who had received at least one dose of melflufen and had an assessment of response (adjusted ITT). This includes 2 patients with a very good partial response (VGPR) and 6 patients with a partial response (PR). Two additional patients achieved minimal response (MR) for a clinical benefit rate (CBR) of 33%.

RESULTS – EFFICACY

Table 5. Overall response rate (N=30)

N	PD	SD	MR	PR	VGPR	ORR	CBR
Adjusted ITT, n (%)	11 (37)	9 (30)	2 (7)	6 (20)	2 (7)	26.7%	33.3%

Figure 1. Waterfall plot (N=30)

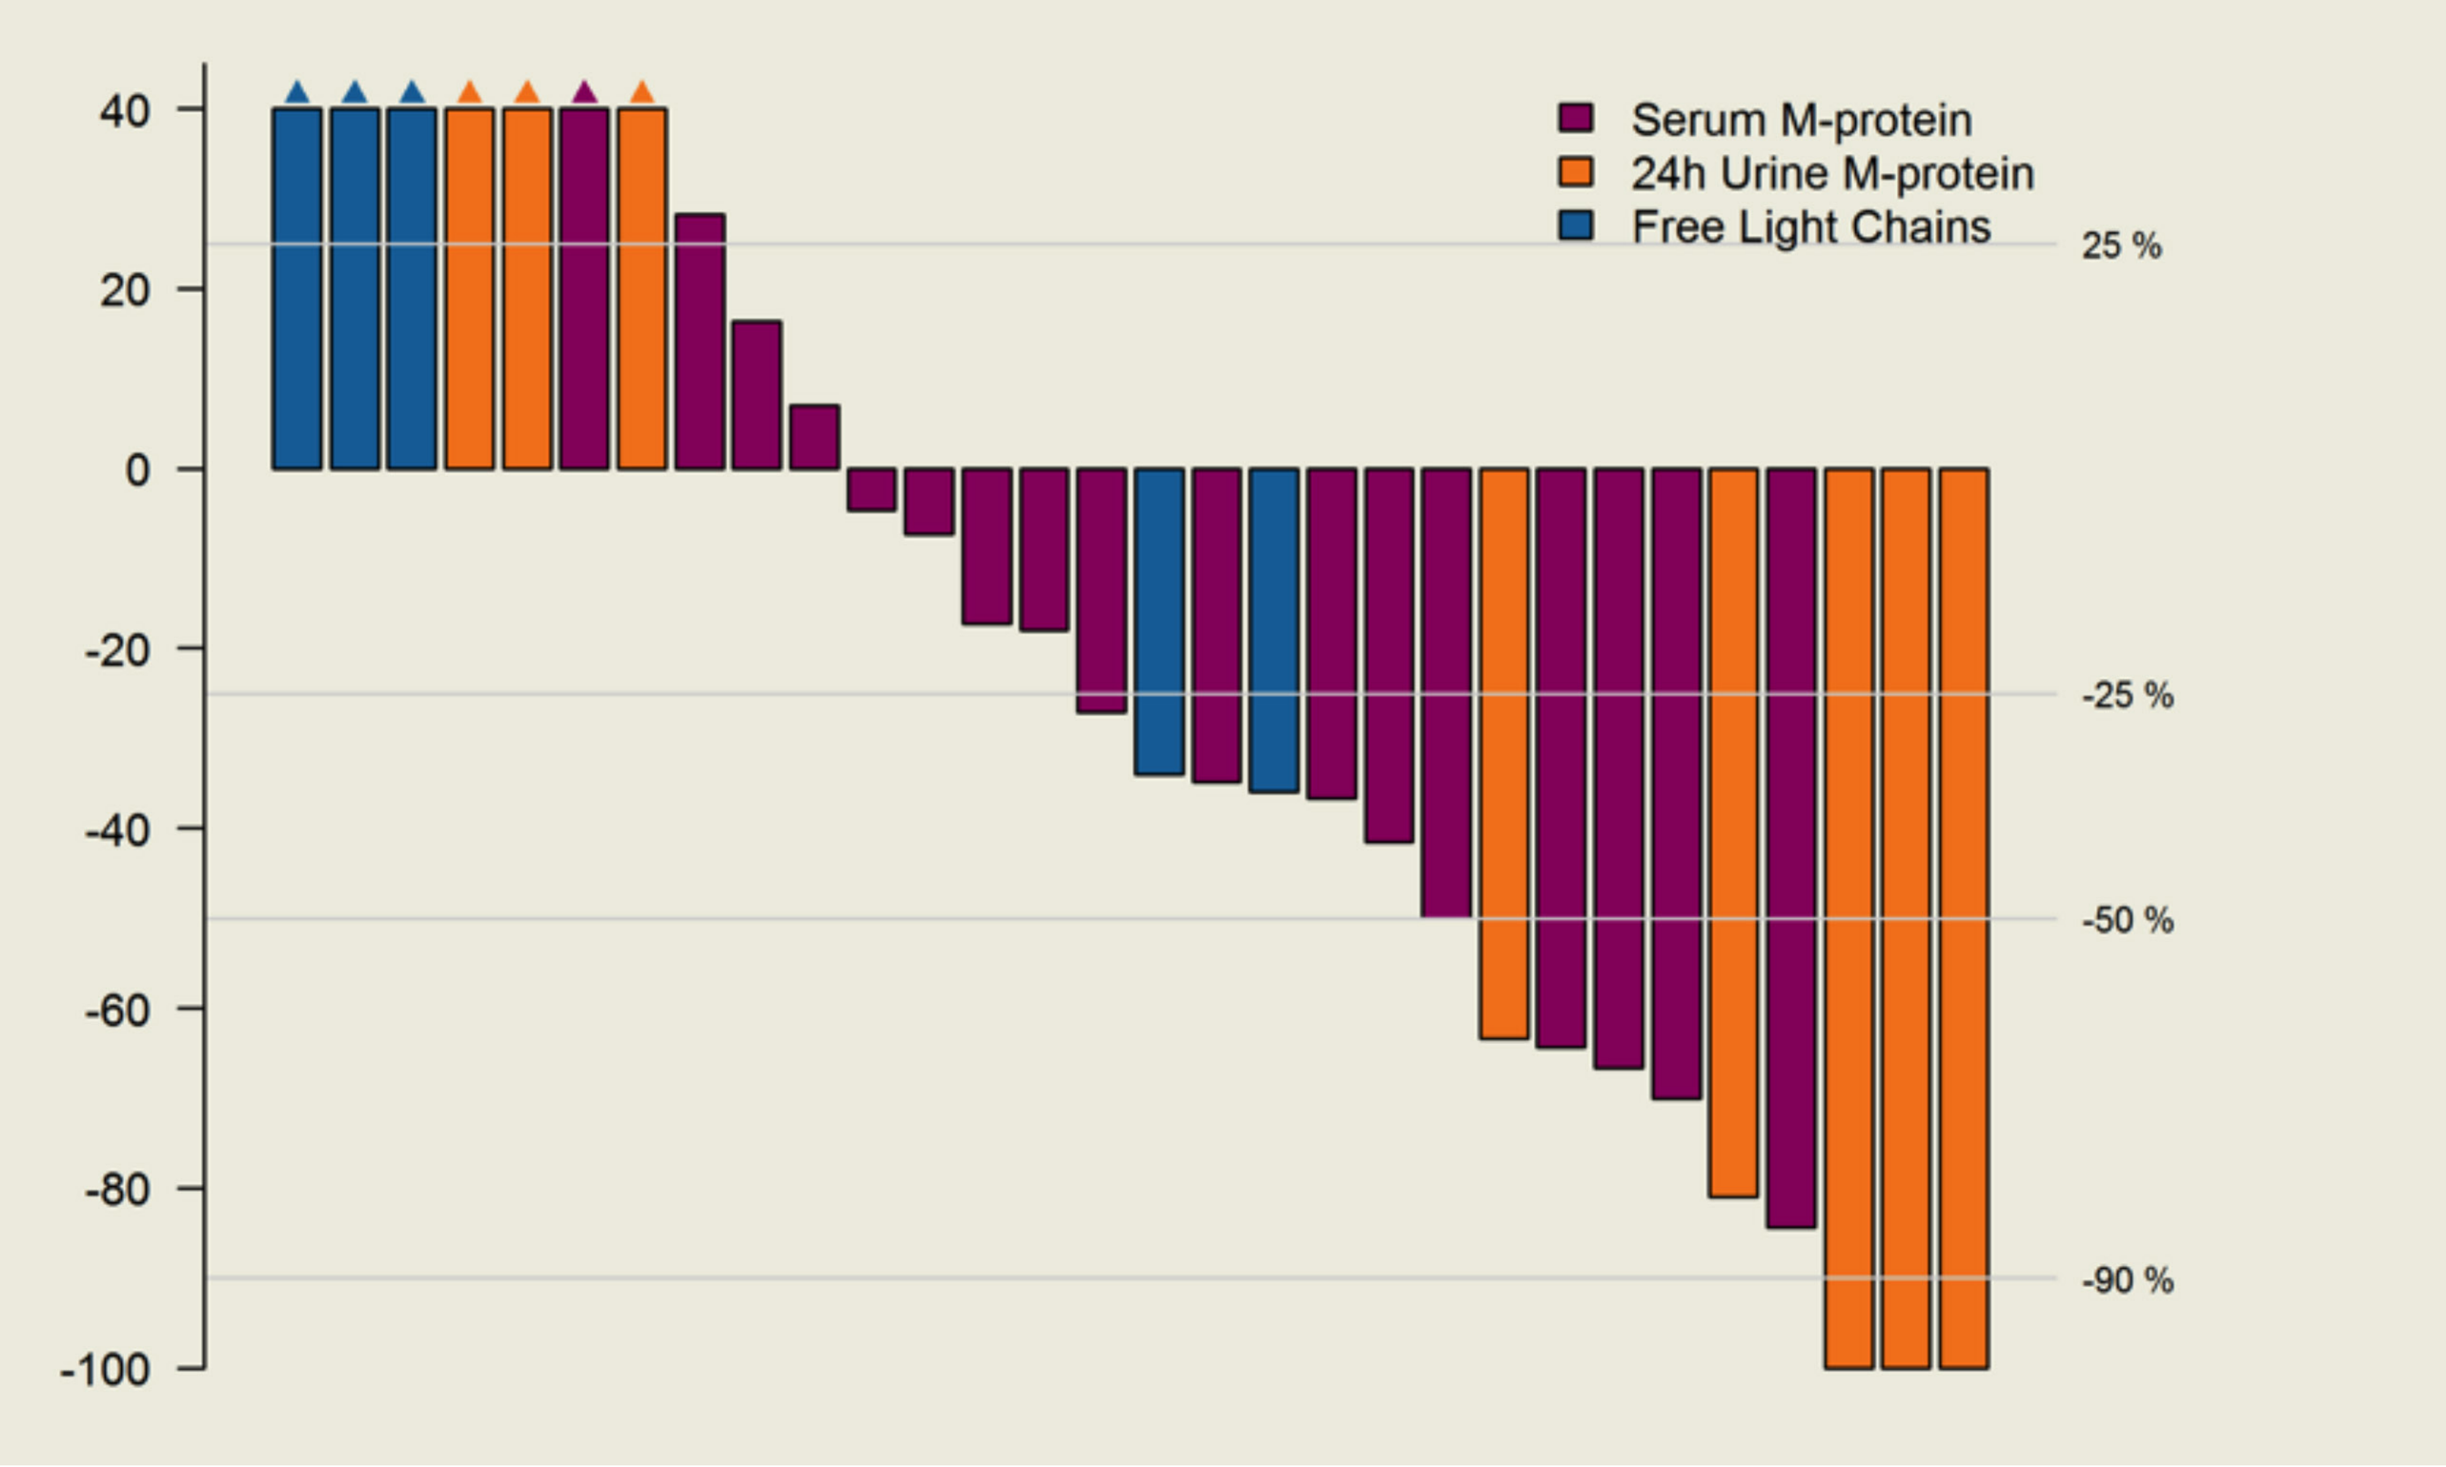
Serum M-protein
24h Urine M-protein
Free Light Chains
40
20
0
-20
-40
-60
-80
-100
25 %
-25 %
-50 %
-90 %

Figure 2. Patient case study

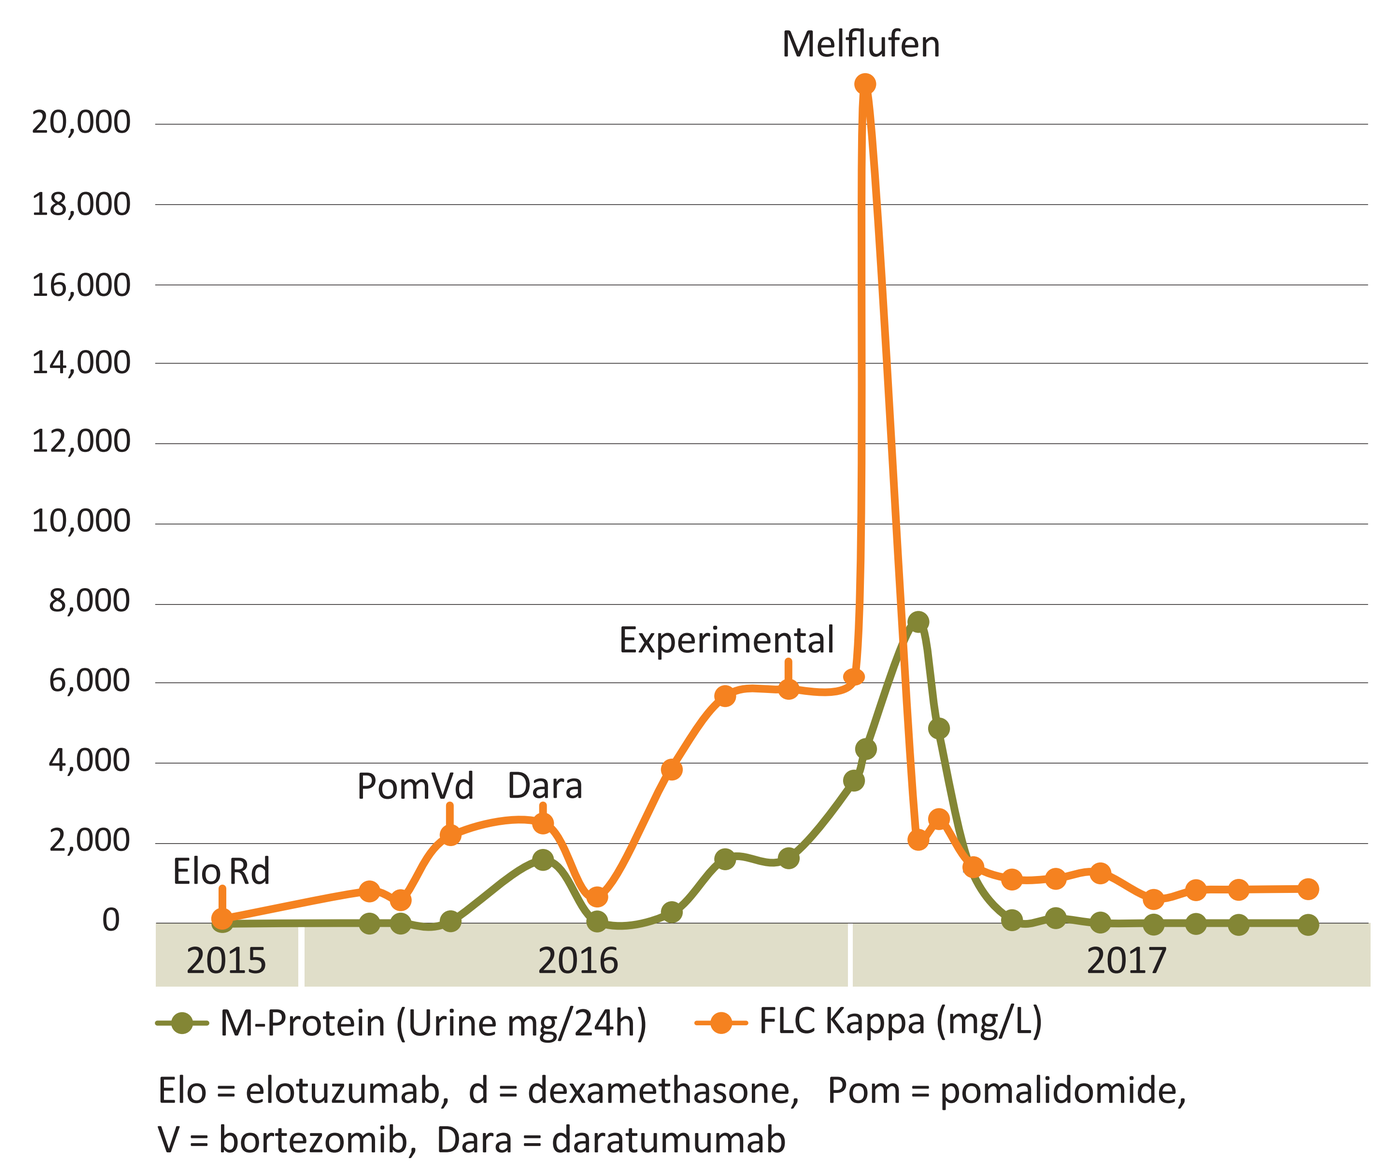
Melflufen
Experimental
PomVd
Dara
Elo Rd
20,000
18,000
16,000
14,000
12,000
10,000
8,000
6,000
4,000
2,000
0
2015
2016
2017
M-Protein (Urine mg/24h)
FLC Kappa (mg/L)

Elo = elotuzumab, d = dexamethasone, Pom = pomalidomide, V = bortezomib, Dara = daratumumab

42-year old man with ISS stage 3, MM diagnosed 2007. No detectable serum M-protein. Nine prior lines of therapy including ASCT X 2 and Allo-SCT. Refractory to R, Elo, V, Pom, Dara and an experimental drug. The patient only achieved PD to the last 4 lines of therapy.

Following 5 cycles of melflufen, the urinary M-protein was undetectable (Figure 2). The patient has received 9 cycles of melflufen, achieved VGPR and is ongoing as of Nov 2017.

Figure 3. Swim-lane plot (N=30)

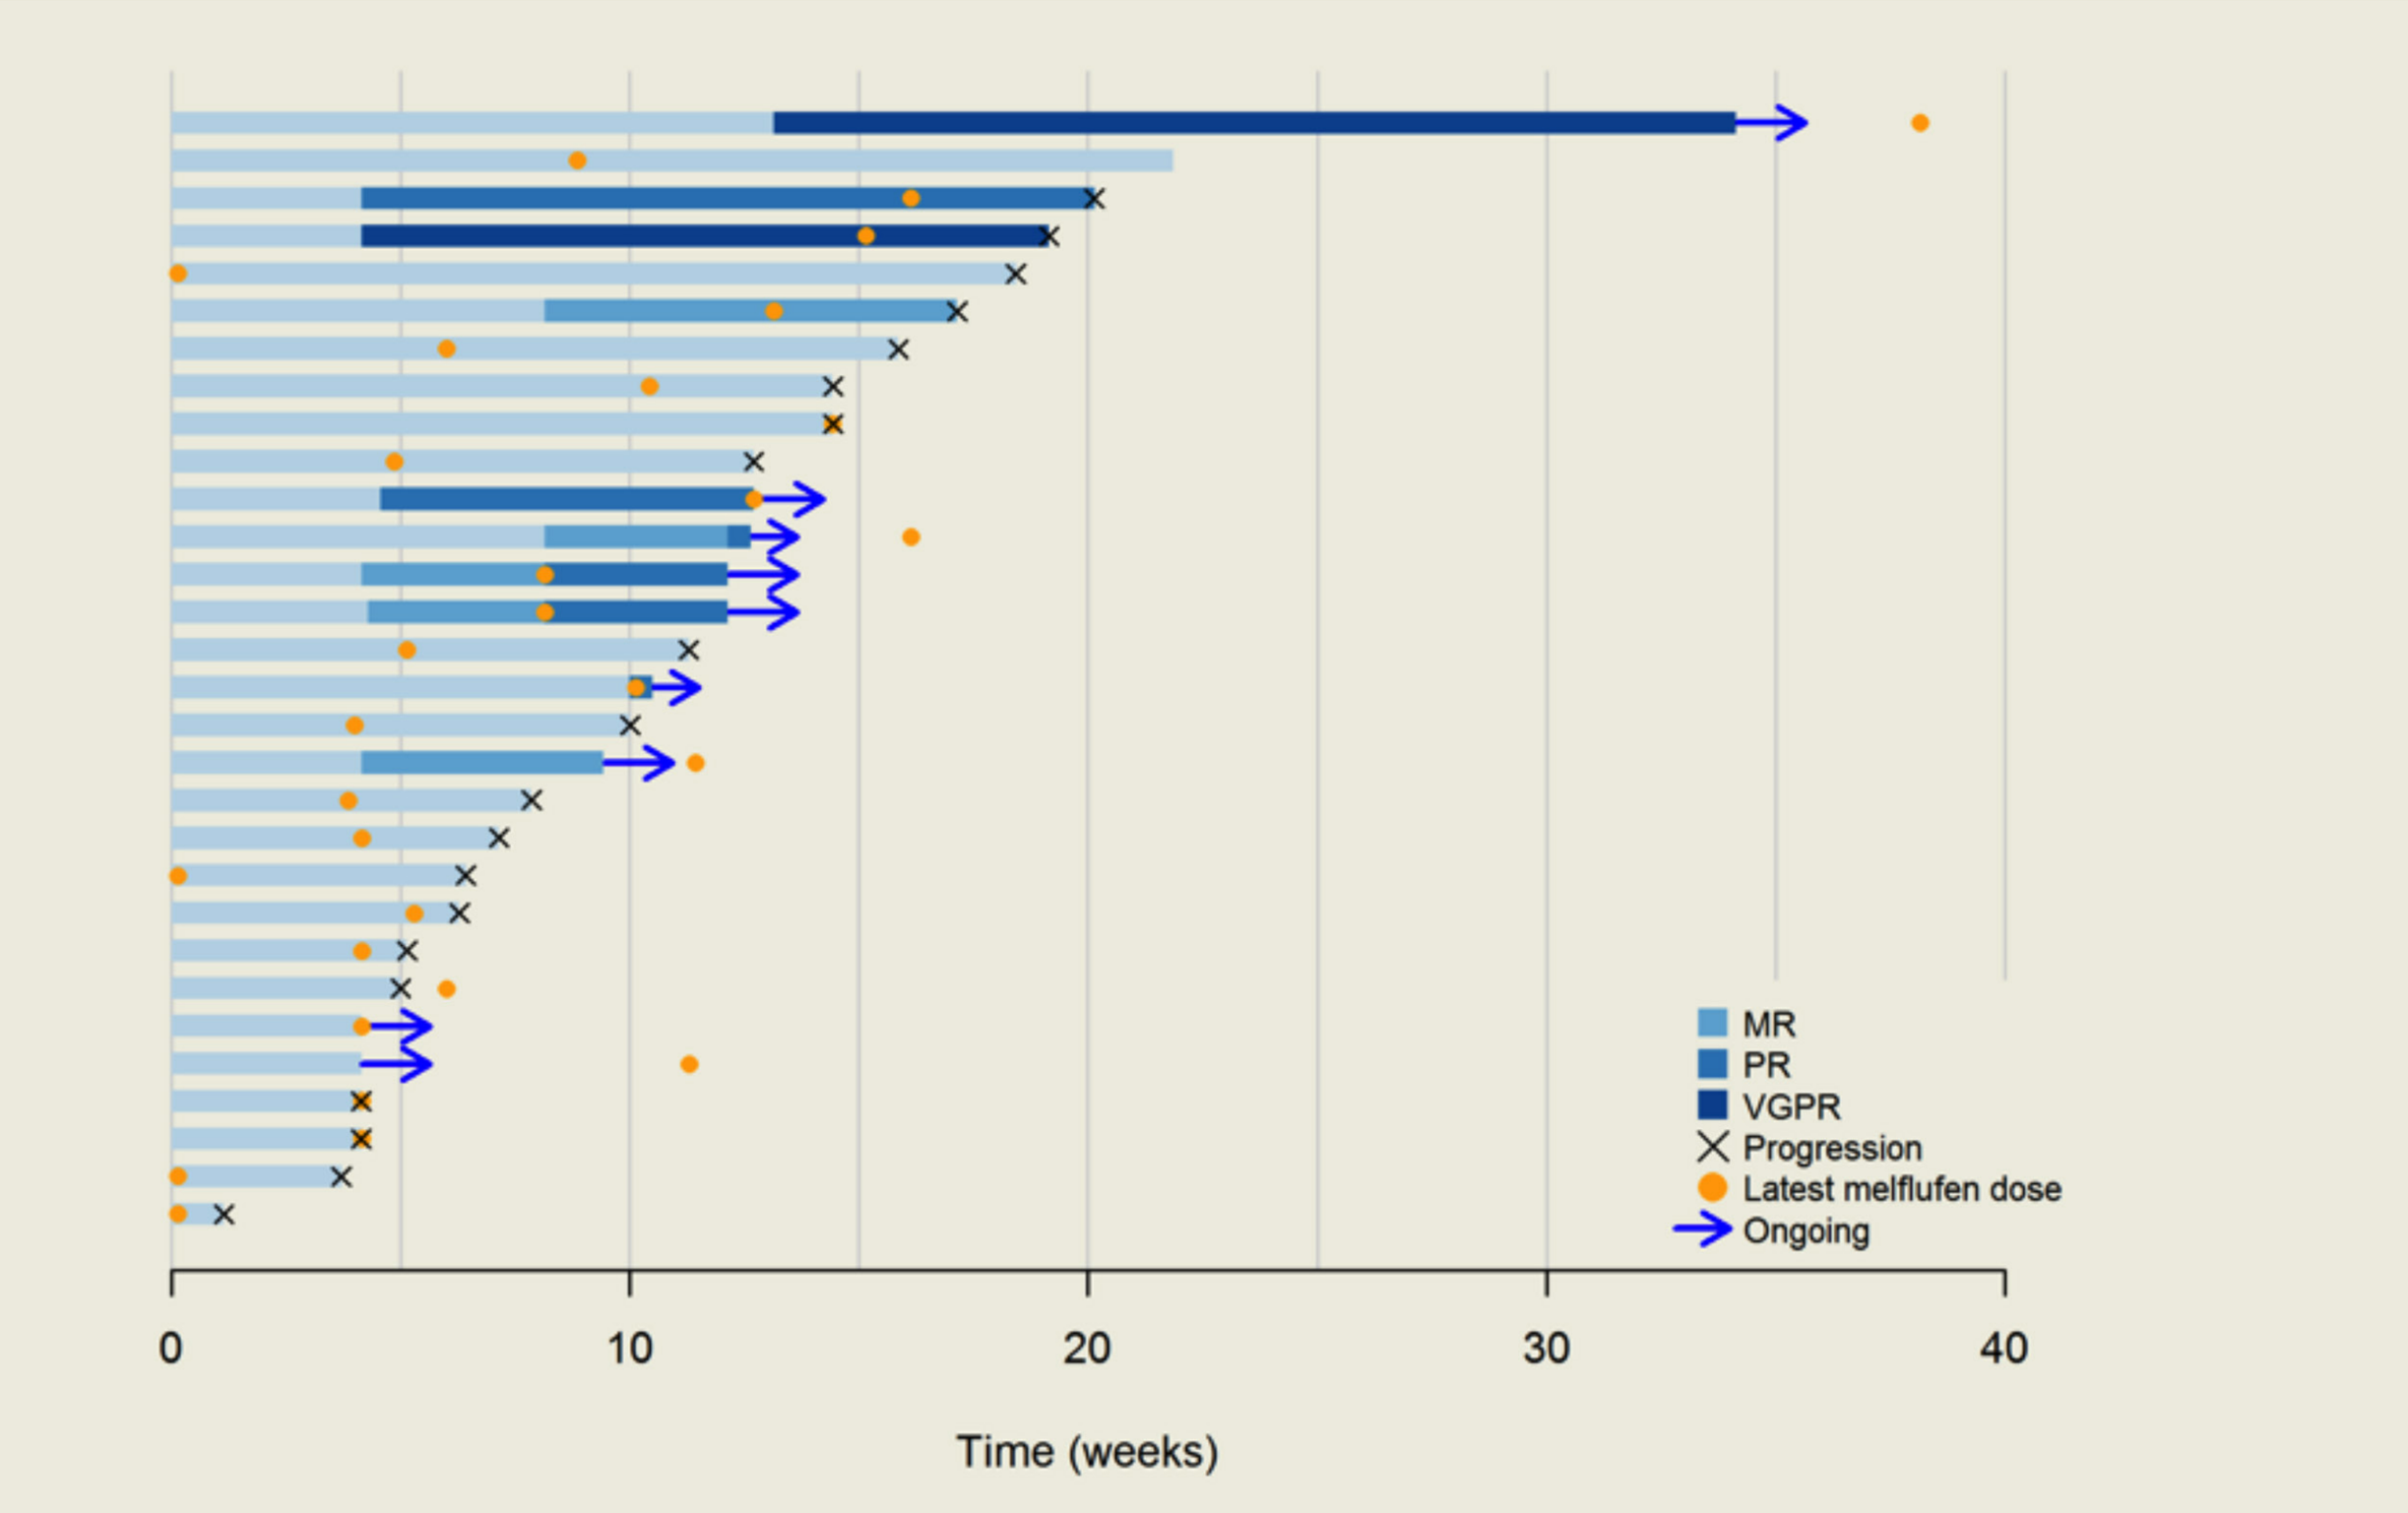
MR
PR
VGPR
Progression
Latest melflufen dose
Ongoing
0
10
20
30
40
Time (weeks)

RESULTS – SAFETY AND TOLERABILITY

Treatment-emergent AEs, regardless of grade, were reported in 35 patients (92%). Treatment-related grade 3/4 AEs were reported in 22 (58%) patients; with those occurring in ≥5% of the patients including thrombocytopenia in 17 (45%) patients, neutropenia in 15 (39%) and anemia in 8 (21%).

Table 6. Treatment-related G3/4 AEs occurring in ≥ 5% of the patients (N=38)

	GRADE 3 OR 4, n (%)	GRADE 4, n (%)
Any treatment-related AE	**22 (58)**	**15 (39)**
Blood and lymphatic system disorders	**20 (53)**	**14 (37)**
Thrombocytopenia	17 (45)	12 (32)
Neutropenia	15 (39)	9 (24)
Leukopenia	3 (8)	3 (8)
Anemia	8 (21)	0
Lymphopenia	3 (8)	0
Hemolytic anemia	2 (5)	0

Eleven patients (35%) experienced a treatment-emergent SAE irrespective of relationship to study treatment. Four patients experienced a melflufen-related SAE (Table 7).

Table 7. Melflufen-related SAEs (N=38)

	n (%)
Any melflufen-related SAE	**4 (11)**
Febrile neutropenia	1 (3)
Pyrexia	1 (3)
Hypercalcemia	1 (3)
Soft tissue infection	1 (3)

CONCLUSION

Following treatment with IMiDs and PIs, patients refractory to pomalidomide and daratumumab have little to no treatment options. Melflufen, a peptidase enhanced therapy with an alkylating payload, demonstrates activity in a heavily refractory population with a median of 6 prior lines of therapy. The efficacy results in this interim analysis are encouraging with an ORR of 27% (including 2 VGPRs) and a CBR of 33%. Melflufen showed a good safety and tolerability profile. Thrombocytopenia and neutropenia were, as expected, the most common AEs, and non-hematologic AEs were infrequent. Melflufen is further evaluated in this ongoing study and the phase 3 study OCEAN (NCT03151811).

ACKNOWLEDGEMENT

The authors would like to thank the patients who volunteered to participate in the study, the staff and the study sites who cared for them, and the CROs involved in data gathering and analyses.

DISCLOSURES

María-Victoria Mateos, Albert Oriol Rocafiguera, Paula Rodríguez Otero, Joan Bladé, Alessandra Larocca, Jan S. Moreb, Michele Cavo, Amitabha Mazumder, Adrián Alegre, Hani Hassoun, Christopher Maisel, Agne Paner, Nashat Gabrail, Kathleen Halka, Jeffrey Zonder, Enrique Ocio and Paul G. Richardson are investigators in the Horizon trial. Paul G. Richardson and María-Victoria Mateos are members of the Oncopeptides AB advisory committee. Catriona Byrne, Johan Harmenberg, Jakob Lindberg, Eva Nordström and Sara Thuresson are working for Oncopeptides AB.